1537P

LEROCICLIB (G1T38), AN ORAL CDK4/6 INHIBITOR, DOSED CONTINUOUSLY IN COMBINATION WITH OSIMERTINIB FOR EGFR^{MUT} NON–SMALL CELL LUNG CANCER: INITIAL PHASE 1B RESULTS

David Berz[1]; Alexander I. Spira[2]; Shirish M. Gadgeel[3]; Ian C. Anderson[4]; Jonathan W. Goldman[5]; Jonathan Thompson[6]; Tracy Foster[4]; Yili Pritchett[7]; Cesar G. Cisneros[7]; Chao Li[7]; Sarika Jain[7]; Jessica A. Sorrentino[7]; Rajesh Malik[7]; and Andrew P. Beelen[7]

[1] Beverly Hills Cancer Center, Beverly Hills, CA, USA; [2] Virginia Cancer Specialists Research Institute, Fairfax, VA, USA; [3] Rogel Cancer Center/University of Michigan, Ann Arbor, MI, USA; [4] St Joseph Heritage Healthcare, Anaheim, CA, USA; [5] David Geffen School of Medicine/UCLA, Los Angeles, CA, USA; [6] Medical College of Wisconsin, Milwaukee, WI, USA; [7] G1 Therapeutics, Research Triangle Park, NC, USA

BACKGROUND

- Epidermal growth factor receptor (EGFR) tyrosine kinase inhibitors (TKIs) are the standard of care for patients with advanced non-small cell lung cancer (NSCLC) with EGFR-activating mutations; however, prognosis remains poor due to acquired resistance that promotes disease recurrence[1]
- Several putative mechanisms of resistance to EGFR TKIs are upstream of the cyclin-dependent kinase 4/6 (CDK4/6) pathway[2]
- Lerociclib (G1T38) is an oral, potent, selective CDK4/6 inhibitor in clinical development
 - Preclinical and early clinical data have demonstrated that lerociclib is differentiated from other CDK4/6 inhibitors based on its favorable safety/tolerability profile and ability to be dosed continuously with less dose-limiting neutropenia (a class effect of CDK4/6 inhibitors)[3,4]
- In *in vitro* and *in vivo* models of NSCLC, lerociclib enhanced efficacy and delayed time to resistance when combined with EGFR TKIs[5,6]
 - Data provide scientific rationale to investigate lerociclib plus osimertinib (TAGRISSO®), a third-generation EGFR TKI, for NSCLC in the clinic
- Based on these data, an open-label, phase 1b/2 study (NCT03455829) was initiated to assess the safety, efficacy, and pharmacokinetics (PK) of lerociclib in combination with osimertinib in patients with EGFR mutation-positive metastatic NSCLC
- The study consists of 2 parts: Part 1 is a phase 1b dose-finding portion. Part 2 is a phase 2 randomized (1:1) portion where patients will receive lerociclib at the recommended phase 2 dose (RP2D) plus osimertinib or osimertinib alone
- Here, we describe preliminary results from the dose escalation part of the study (Part 1)

STUDY OBJECTIVES

Primary objectives

- Evaluate the safety, tolerability, and dose-limiting toxicities (DLTs) of lerociclib administered with osimertinib
- Determine the RP2D of lerociclib administered with osimertinib

Secondary objectives

- Assess the effect of osimertinib on the PK parameters of lerociclib
- Assess objective response rate (ORR), clinical benefit rate (CBR), progression-free survival (PFS), and overall survival (OS)

Key exploratory objectives

- Assess the relationships between efficacy parameters and PK parameters
- Assess correlation between changes in cell-free DNA (cfDNA) with clinical outcomes
- Assess correlation between NSCLC subtype and clinical outcomes

METHODS

Patients

- Key eligibility criteria:
 - Age ≥ 18 years
 - Histologically or cytologically confirmed diagnosis of NSCLC (stage IV)
 - Known EGFR mutation associated with EGFR TKI sensitivity
 - Prior treatment with ≤ 2 lines of chemotherapy in the advanced setting
 - Eastern Cooperative Oncology Group performance status: 0 or 1
 - Adequate organ function

Study Design

- Part 1 (phase 1b): dose escalation (**Figure 1**)
 - Patients received escalating doses of oral lerociclib on a once daily (QD) or twice daily (BID) continuous schedule in combination with osimertinib 80 mg QD until disease progression or unacceptable toxicity
 - The planned QD doses for lerociclib were 200, 300, 400, 500, and 650 mg using a standard 3 + 3 escalation method based on occurrence of DLTs in cycle 1 to determine the lerociclib RP2D
 - Based on data from another ongoing study (NCT02983071), the lerociclib 500 mg QD and 650 mg QD doses were not explored in this study; lerociclib 150 mg BID and 200 mg BID doses were also investigated
 - DLTs were defined as any of the following drug-related adverse events (AEs):
 - Grade 4 neutropenia
 - Grade ≥ 3 neutropenic infection/febrile neutropenia
 - Grade 4 thrombocytopenia
 - Grade ≥ 3 thrombocytopenia with bleeding
 - Grade ≥ 3 nonhematologic toxicity
 - Liver function test abnormalities meeting Hy's Law criteria

Figure 1. Study Schematic for Part 1

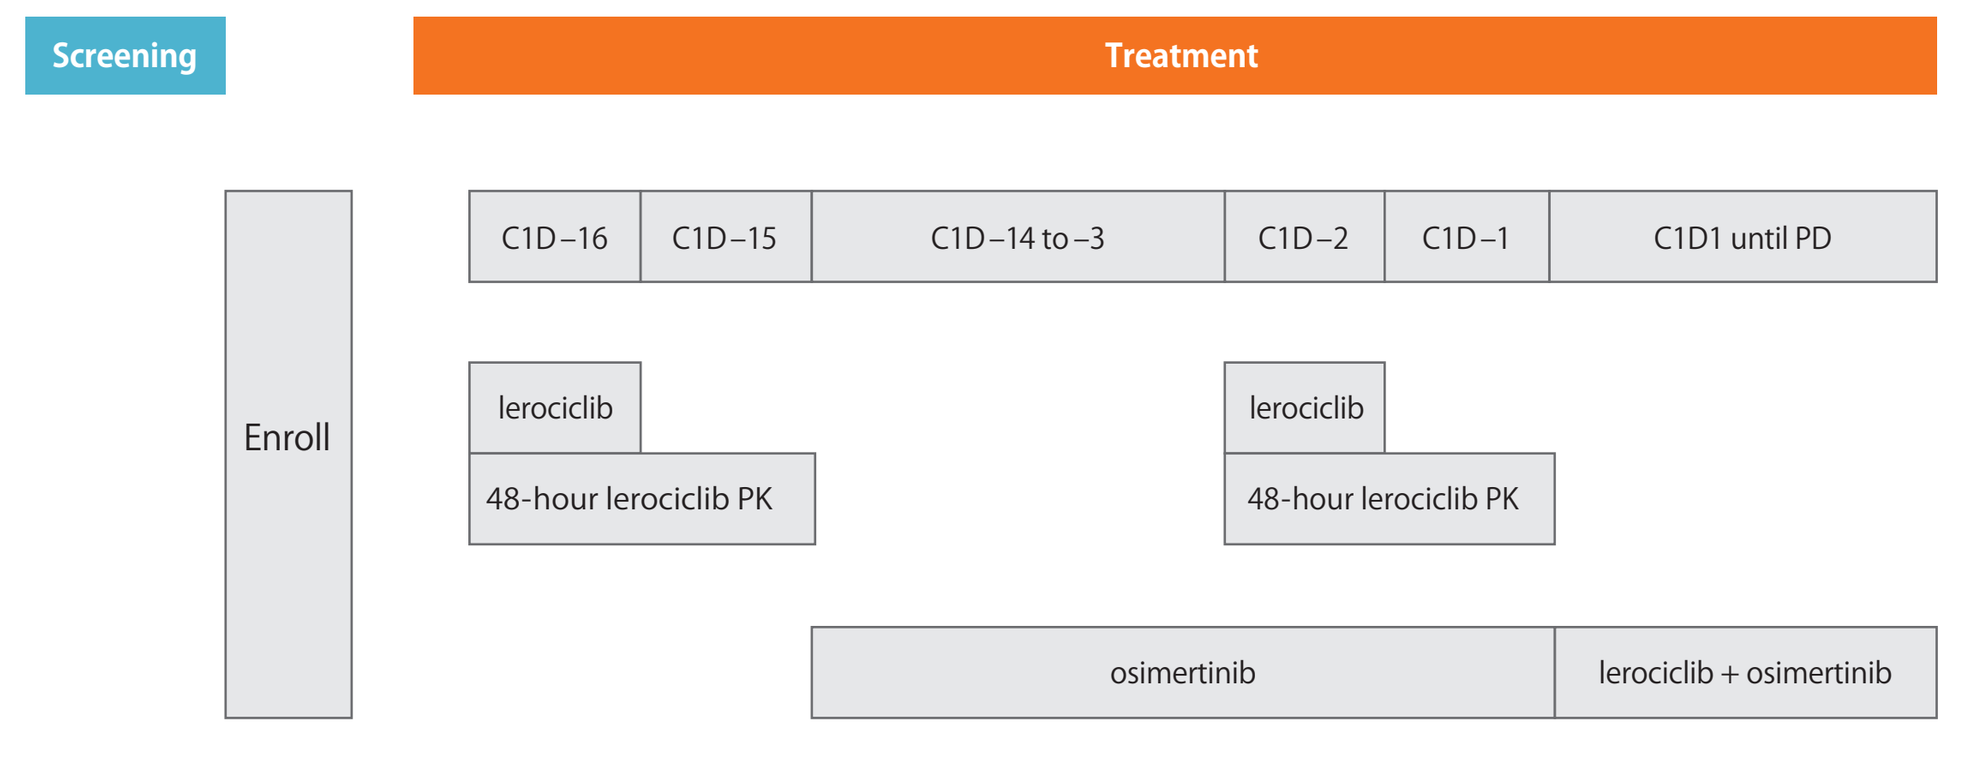
Screening
Treatment
PTV
Survival
Enroll
C1D−16
C1D−15
C1D−14 to −3
C1D−2
C1D−1
C1D1 until PD
lerociclib
48-hour lerociclib PK
lerociclib
48-hour lerociclib PK
osimertinib
lerociclib + osimertinib
All patients assessed radiographically every 8 weeks until disease progression, then via telephone every 3 months for long-term survival

1 cycle = 28 days.
C, cycle; D, day; PD, progressive disease; PK, pharmacokinetics; PTV, post-treatment visit.

Assessments

- AEs were graded according to the National Cancer Institute's Common Terminology Criteria for AEs version 4.03
- Tumor response was assessed per Response Evaluation Criteria in Solid Tumors version 1.1 (RECIST v1.1) based on the response evaluable patient population, defined as treated patients who had a measurable target lesion at baseline and at least 1 post baseline tumor assessment, had clinical PD, or died before the first post baseline tumor assessment
- PK blood samples were collected on cycle 1 day −16 (pre dose and up to 48 hours post lerociclib administration) and cycle 1 day −2 (coadministered with osimertinib; pre dose and up to 48 hours post lerociclib administration)
 - PK parameters (including time to maximum concentration [T_{max}], maximum concentration [C_{max}], area under the curve from time 0 to time of last measurable concentration [AUC_{last}], and area under the curve from time 0 to infinity [$AUC_{0\text{-}inf}$]) were determined on cycle 1 day −16 and day −2 by noncompartmental analysis (Phoenix® software) for lerociclib and its pharmacologically active metabolite (G1T30)
- Blood samples for cfDNA analysis were collected at baseline and on day 15 of cycle 1, then day 1 of odd-numbered cycles, starting with cycle 3. After 18 months of treatment, subsequent cfDNA sample collection was performed every 12 weeks until disease progression. Samples were analyzed at Inivata Ltd.

RESULTS

Patient Disposition and Baseline Characteristics

- As of August 01, 2019, 26 patients have been enrolled; 13 patients remain on study treatment
- Reasons for treatment discontinuation were disease progression (n = 12) and investigator's discretion (n = 1)
- Patient demographic and baseline disease characteristics are summarized in **Table 1**

Table 1. Patient Demographics and Baseline Disease Characteristics

	200 mg QD (n = 7)	300 mg QD (n = 6)	400 mg QD (n = 4)	150 mg BID (n = 3)	200 mg BID (n = 6)	Overall (N = 26)
Median (range) age, years	59.0 (53–87)	64.5 (50–73)	60.5 (56–74)	47.0 (44–73)	65.5 (62–71)	62.0 (44–87)
Male, n (%)	1 (14.3)	3 (50.0)	1 (25.0)	1 (33.3)	2 (33.3)	8 (30.8)
Race, n (%)						
Asian	1 (14.3)	1 (16.7)	1 (25.0)	0	1 (16.7)	4 (15.4)
Black or African American	0	0	0	2 (66.7)	1 (16.7)	3 (11.5)
White	1 (14.3)	5 (83.3)	2 (50.0)	1 (33.3)	4 (66.7)	13 (50.0)
Other	5 (71.4)	0	1 (25.0)	0	0	6 (23.1)
ECOG PS, n (%)						
0	3 (42.9)	3 (50.0)	0	3 (100.0)	5 (83.3)	14 (53.8)
1	4 (57.1)	3 (50.0)	4 (100.0)	0	1 (16.7)	12 (46.2)
Histopathologic type at initial diagnosis, n (%)						
Adenocarcinoma	7 (100.0)	5 (83.3)	4 (100.0)	3 (100.0)	5 (83.3)	24 (92.3)
Squamous cell carcinoma	0	1 (16.7)	0	0	1 (16.7)	2 (7.7)
Identified EGFR mutation, n (%)						
Exon 19 deletion	6 (85.7)	6 (100.0)	2 (50.0)	2 (66.7)	3 (50.0)	19 (73.1)
L858R	1 (14.3)	1 (16.7)	1 (25.0)	0	2 (33.3)	5 (19.2)
L861Q	0	0	1 (25.0)	0	0	1 (3.8)
T790M	1 (14.3)	1 (16.7)	0	1 (33.3)	3 (50.0)	6 (23.1)
Other[a]	1 (14.3)	0	0	0	2 (33.3)	3 (11.5)
History of smoking, n (%)	1 (14.3)	3 (50.0)	1 (25.0)	0	3 (50.0)	8 (30.8)
Prior therapy in advanced/metastatic setting, n (%)						
None	0	1 (16.7)	1 (25.0)	1 (33.3)	0	3 (11.5)
Chemotherapy	2 (28.6)	3 (50.0)	2 (50.0)	1 (33.3)	3 (50.0)	11 (42.3)
EGFR TKI[b]	7 (100.0)	4 (66.7)	3 (75.0)	2 (66.7)	5 (83.3)	21 (80.8)
Osimertinib	4 (57.1)	4 (66.7)	3 (75.0)	2 (66.7)	4 (66.7)	17 (65.4)
Immunotherapy[c]	0	2 (33.3)	1 (25.0)	1 (33.3)	2 (33.3)	6 (23.1)
Other	2 (28.6)	0	1 (25.0)	1 (33.3)	1 (16.7)	5 (19.2)

[a] Includes the following EGFR/other mutations: A871G, P1326Q, P1325P, K267, N399FS, exon 20 insertion, TP53 L252_I254.
[b] Erlotinib, gefitinib, afatinib, osimertinib, tesevatinib, HS-10296, or ASP8273.
[c] Atezolizumab, nivolumab, or pembrolizumab.
BID, twice daily; ECOG PS, Eastern Cooperative Oncology Group performance status; EGFR, epidermal growth factor receptor; QD, once daily; TKI, tyrosine kinase inhibitor.

Safety and Tolerability

- The maximum tolerated dose (MTD) has not been defined
- 1 of 3 patients experienced a DLT of Grade 4 neutropenia at 400 mg QD
- The 400 mg QD cohort was expanded with 3 additional patients at 200 mg BID and no additional DLTs were observed
- An additional cohort was enrolled at 150 mg BID to further define the optimal dose
- Lerociclib dose reduction occurred in 3 patients (200, 300, and 400 mg QD dose levels)
- The most common treatment emergent AEs (TEAEs) (≥ 20% of all patients) are reported in **Table 2**
- Only 1 (3.8%) Grade 4 lerociclib-related TEAE was reported (neutropenia)
- No serious treatment-related AEs have been reported to date
- There were no reported AEs of venous thromboembolism (VTE), QT prolongation, or drug-induced liver injury (DILI)
- There were no AEs leading to lerociclib/osimertinib discontinuation or death

Table 2. Most Common TEAEs (≥ 20% of All Patients)

Patients, n (%)	200 mg QD (n = 7)[a]		300 mg QD (n = 6)		400 mg QD (n = 4)[a]		150 mg BID (n = 3)		200 mg BID (n = 6)		Overall (N = 26)	
Grade	All	≥ 3	All	≥ 3	All	≥ 3	All	≥ 3	All	≥ 3	All	≥ 3
Any AE	7 (100.0)	4 (57.1)[b]	6 (100.0)	3 (50.0)	4 (100.0)	3 (75.0)[b]	3 (100.0)	0	4 (66.7)	2 (33.3)[b]	24 (92.3)	12 (46.2)
Diarrhea	4 (57.1)	2 (28.6)	6 (100.0)	1 (16.7)	4 (100.0)	0	3 (100.0)	0	2 (33.3)	1 (16.7)	19 (73.1)	4 (15.4)
Neutropenia	6 (85.7)	1 (14.3)	4 (66.7)	0	3 (75.0)	2 (50.0)[b]	1 (33.3)	0	0	0	14 (53.8)	3 (11.5)[b]
Nausea	3 (42.9)	0	5 (83.3)	0	3 (75.0)	0	1 (33.3)	0	1 (16.7)	0	13 (50.0)	0
Vomiting	4 (57.1)	0	4 (66.7)	0	2 (50.0)	0	1 (33.3)	0	1 (16.7)	0	12 (46.2)	0
Leukopenia	5 (71.4)	1 (14.3)	3 (50.0)	1 (16.7)	1 (25.0)	1 (25.0)	1 (33.3)	0	0	0	10 (38.5)	3 (11.5)
Anemia	4 (57.1)	1 (14.3)	2 (33.3)	0	2 (50.0)	0	0	0	0	0	8 (30.8)	1 (3.8)
Cough	2 (28.6)	0	1 (16.7)	0	2 (50.0)	0	1 (33.3)	0	1 (16.7)	0	7 (26.9)	0
Headache	4 (57.1)	0	1 (16.7)	0	1 (25.0)	0	1 (33.3)	0	0	0	7 (26.9)	0
Lymphocytopenia	5 (71.4)	1 (14.3)	1 (16.7)	0	1 (25.0)	1 (25.0)	0	0	0	0	7 (26.9)	2 (7.7)
Thrombocytopenia	1 (14.3)	0	4 (66.7)	0	2 (50.0)	1 (25.0)	0	0	0	0	7 (26.9)	1 (3.8)
Decreased appetite	2 (28.6)	0	2 (33.3)	0	1 (25.0)	0	1 (33.3)	0	0	0	6 (23.1)	0
Hyperglycemia	4 (57.1)	0	1 (16.7)	1 (16.7)	0	0	1 (33.3)	0	0	0	6 (23.1)	1 (3.8)

BID, twice daily; QD, once daily; TEAE, treatment emergent adverse event.
[a] 1 patient was replaced due to discontinuation (disease progression) prior to the end of the DLT period.
[b] 1 patient with a Grade 4 event.

Neutrophil Effects

- Continuous lerociclib dosing with osimertinib resulted in a dose-dependent decline and subsequent plateau of neutrophils at week 4 (**Figure 2**)

Figure 2. Median Neutrophils (Absolute) by Visits

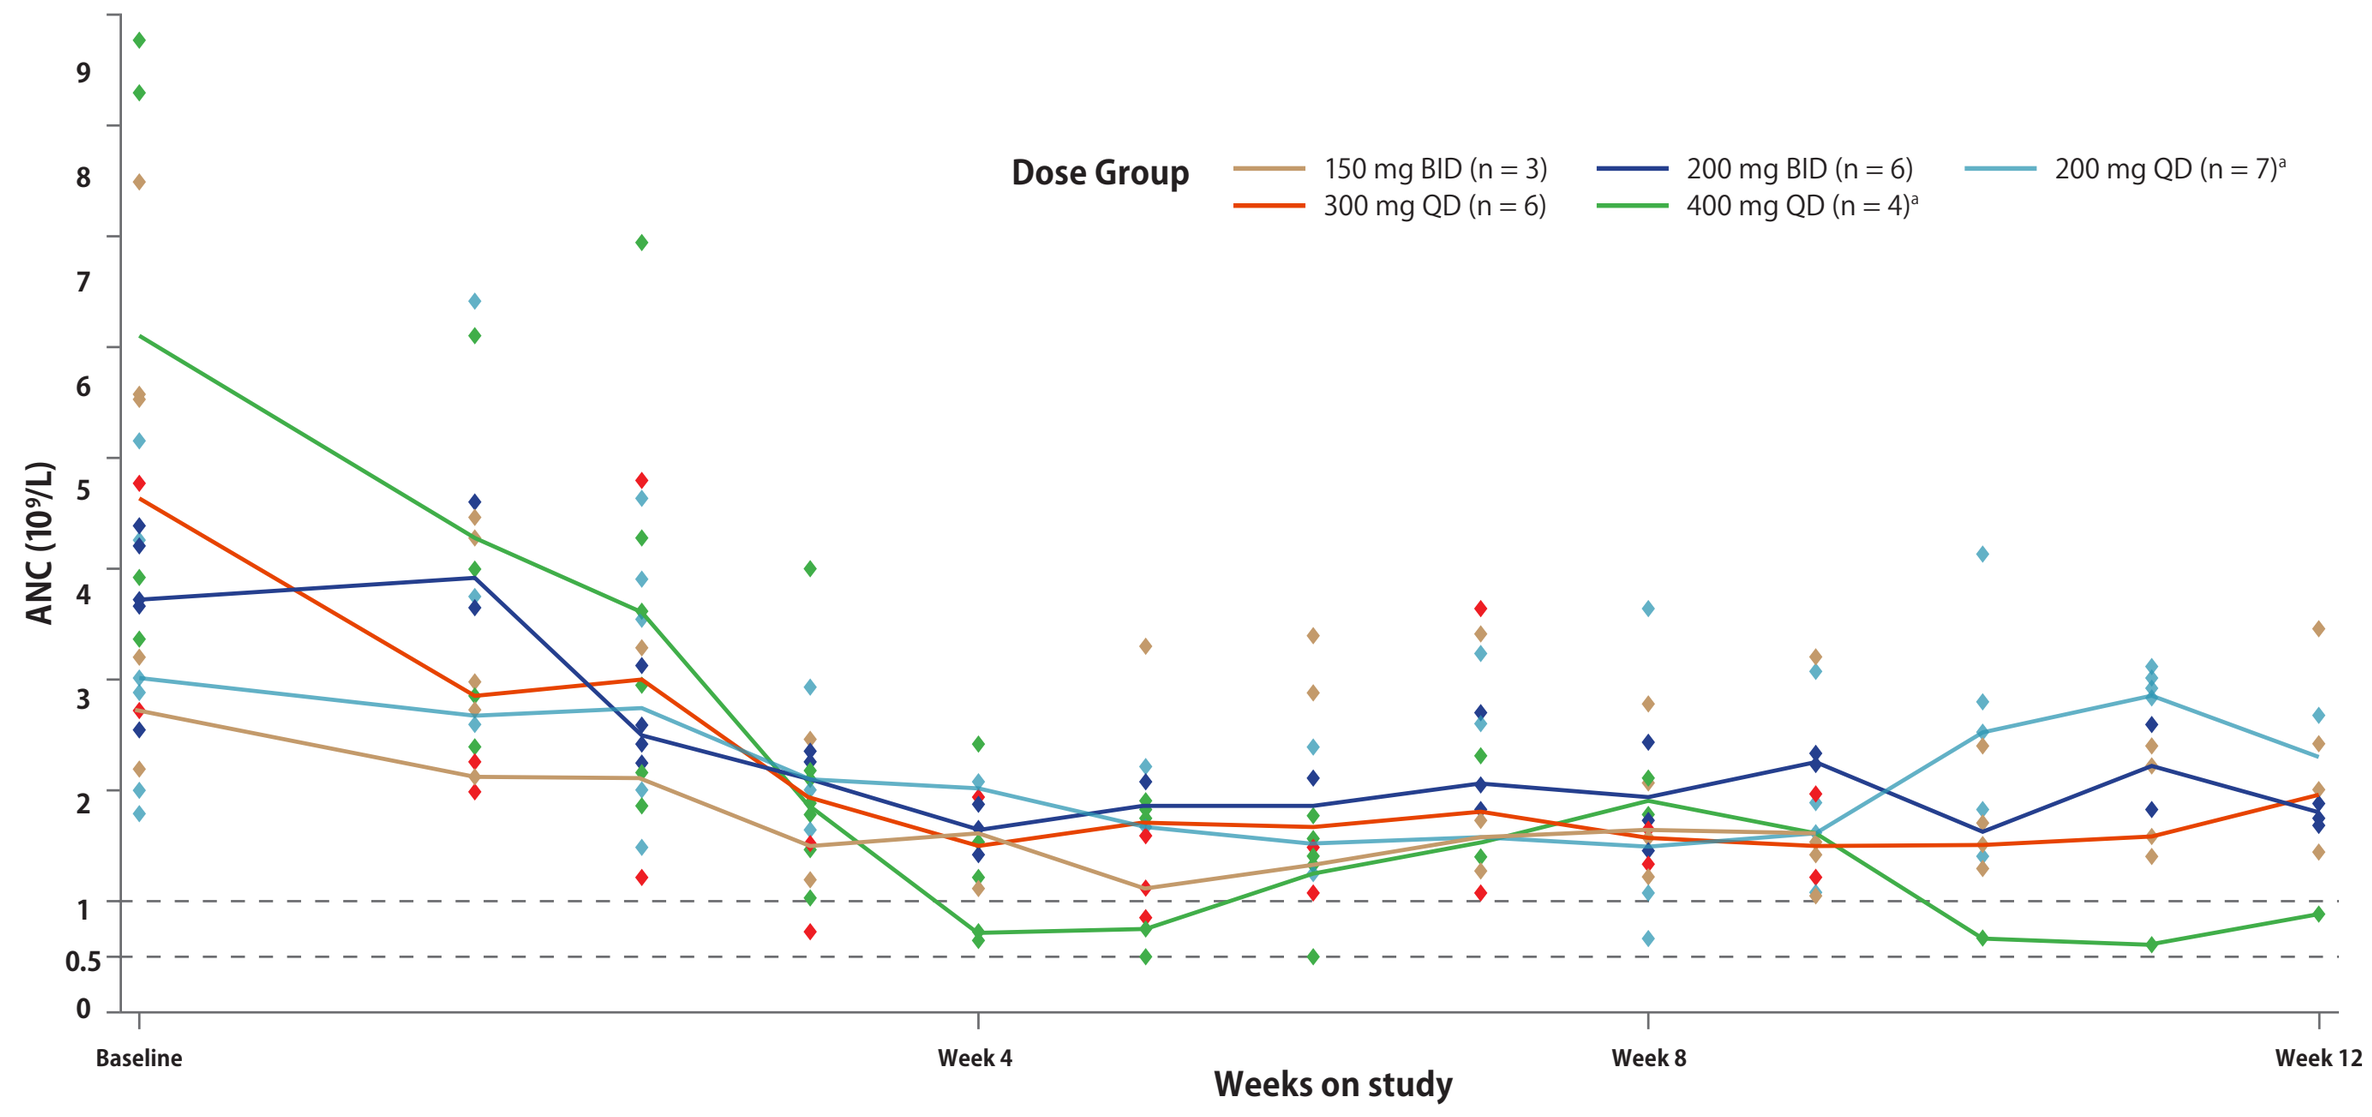
Dose Group
150 mg BID (n = 3)
300 mg QD (n = 6)
200 mg BID (n = 6)
400 mg QD (n = 4)[a]
200 mg QD (n = 7)[a]
ANC (10⁹/L)
Baseline
Week 4
Week 8
Week 12
Weeks on study

Baseline is defined as the last available value prior to or on the date in the PK period.
[a] 1 patient was replaced due to discontinuation (disease progression) prior to the end of the DLT period.
ANC, absolute neutrophil count; BID, twice daily; QD, once daily.

Efficacy

- 3 confirmed objective partial responses were observed in osimertinib-naïve patients at the 200 mg QD (n = 2) and 300 mg QD (n = 1) dose levels (**Table 3; Figure 3** and **Figure 4**)
- 2 patients had an unconfirmed partial response (1 patient is still on treatment [150 mg BID] and has not reached confirmatory scan time point)

Table 3. Best Overall Response (Confirmed)

Patients, n (%)	200 mg QD (n = 7)	300 mg QD (n = 6)	400 mg QD (n = 4)	150 mg BID (n = 3)	200 mg BID (n = 3)	Overall (N = 23)
Best overall response						
CR	0	0	0	0	0	0
PR	2 (28.6)	1 (16.7)	0	0	0	3 (13.0)
SD[a]	2 (28.6)	4 (66.7)	1 (25.0)	2 (66.7)	2 (66.7)	11 (47.8)
PD	1 (14.3)	1 (16.7)	3 (75.0)	1 (33.3)	0	6 (26.1)
NE	2 (28.6)	0	0	0	1 (33.3)	3 (13.0)[b]
ORR	2 (28.6)	1 (16.7)	0	0	0	3 (13.0)
CR + PR + SD ≥ 8 weeks	4 (57.1)	5 (83.3)	1 (25.0)	2 (66.7)	2 (66.7)	14 (60.9)

[a] Includes 2 patients with unconfirmed PR in 150 mg BID and 200 mg BID groups.
[b] 2 patients discontinued treatment due to clinical PD before the first scheduled tumor scan. 1 patient discontinued treatment due to radiographic PD with incomplete tumor assessment data.
CR, complete response; NE, not evaluable; ORR, objective response rate (CR + PR); PD, progressive disease; PR, partial response; SD, stable disease.
8 weeks = 56 days +/- 7 days.

Figure 3. Treatment Duration and Response by Dose Group

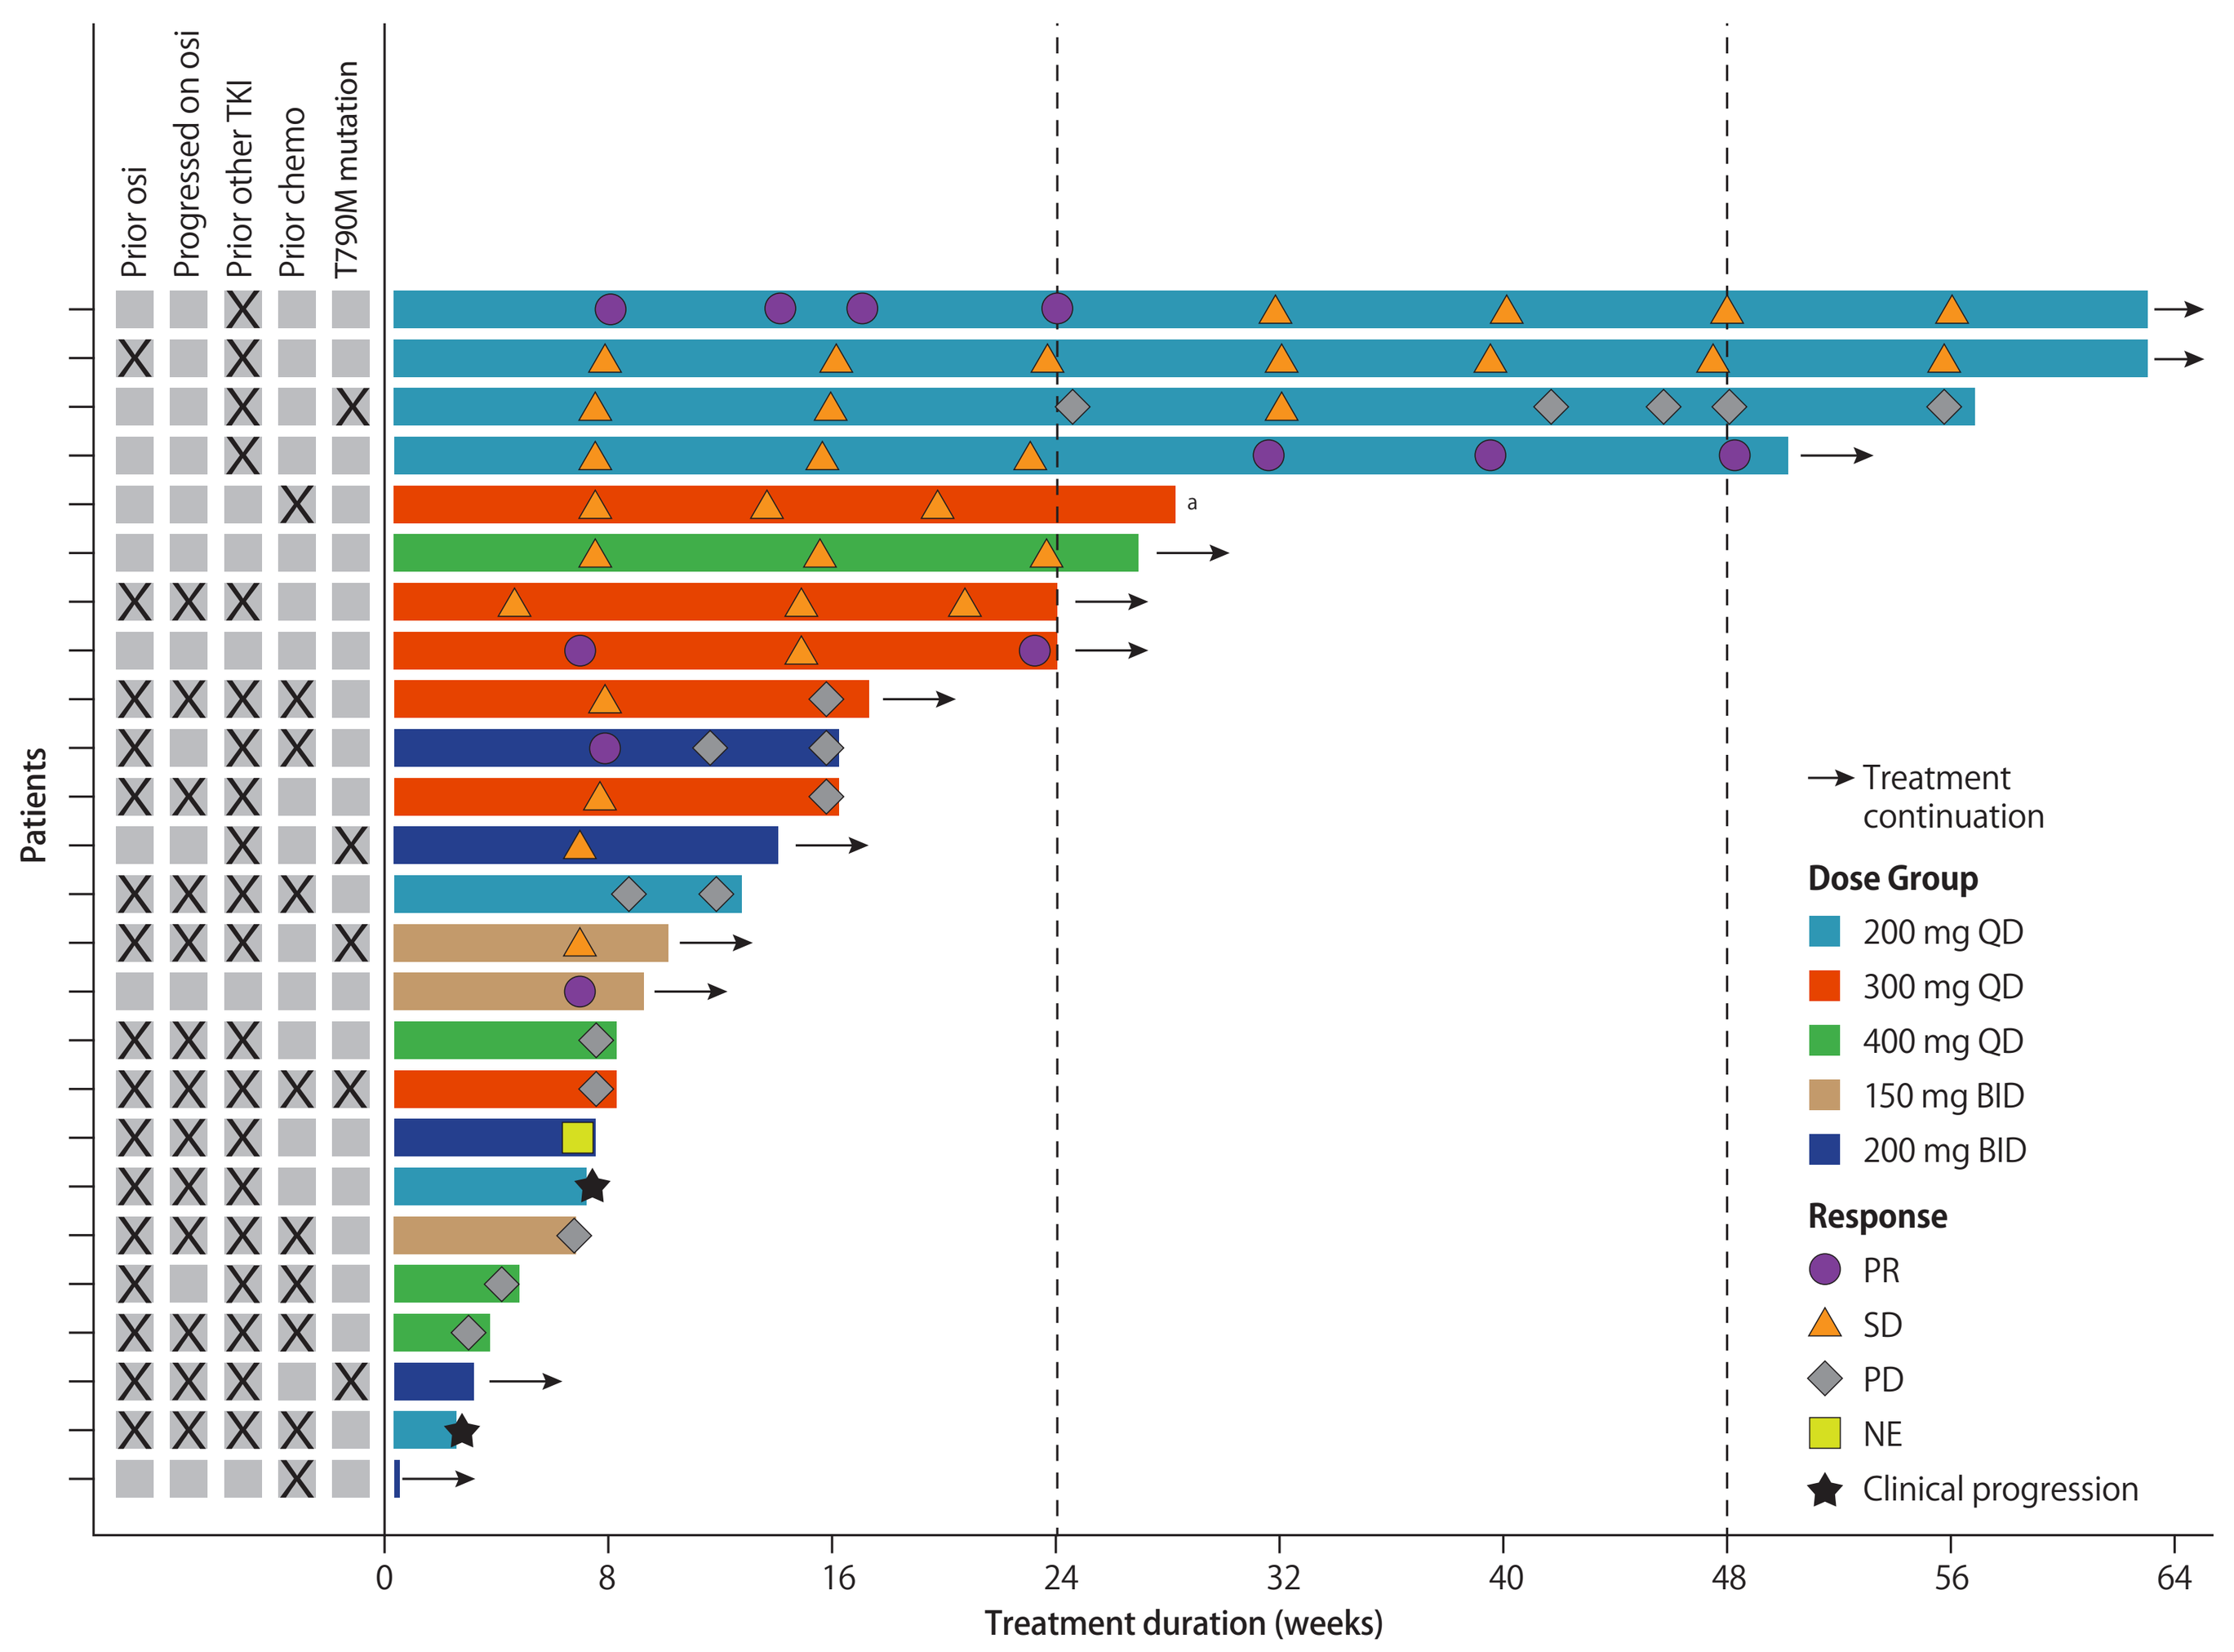
Prior osi
Progressed on osi
Prior other TKI
Prior chemo
T790M mutation
Patients
Treatment continuation
Dose Group
200 mg QD
300 mg QD
400 mg QD
150 mg BID
200 mg BID
Response
PR
SD
PD
NE
Clinical progression
Treatment duration (weeks)

Patients who started the continuous daily dosing (cycle 1 day 1) were included (n = 25).
[a] 1 patient discontinued treatment due to investigator's discretion.
BID, twice daily; chemo, chemotherapy; NE, not evaluable; osi, osimertinib; PD, progressive disease; PR, partial response; QD, once daily; SD, stable disease; TKI, tyrosine kinase inhibitor.

Figure 4. Best Relative Change From Baseline in Tumor Size for Target Lesions by Dose Level

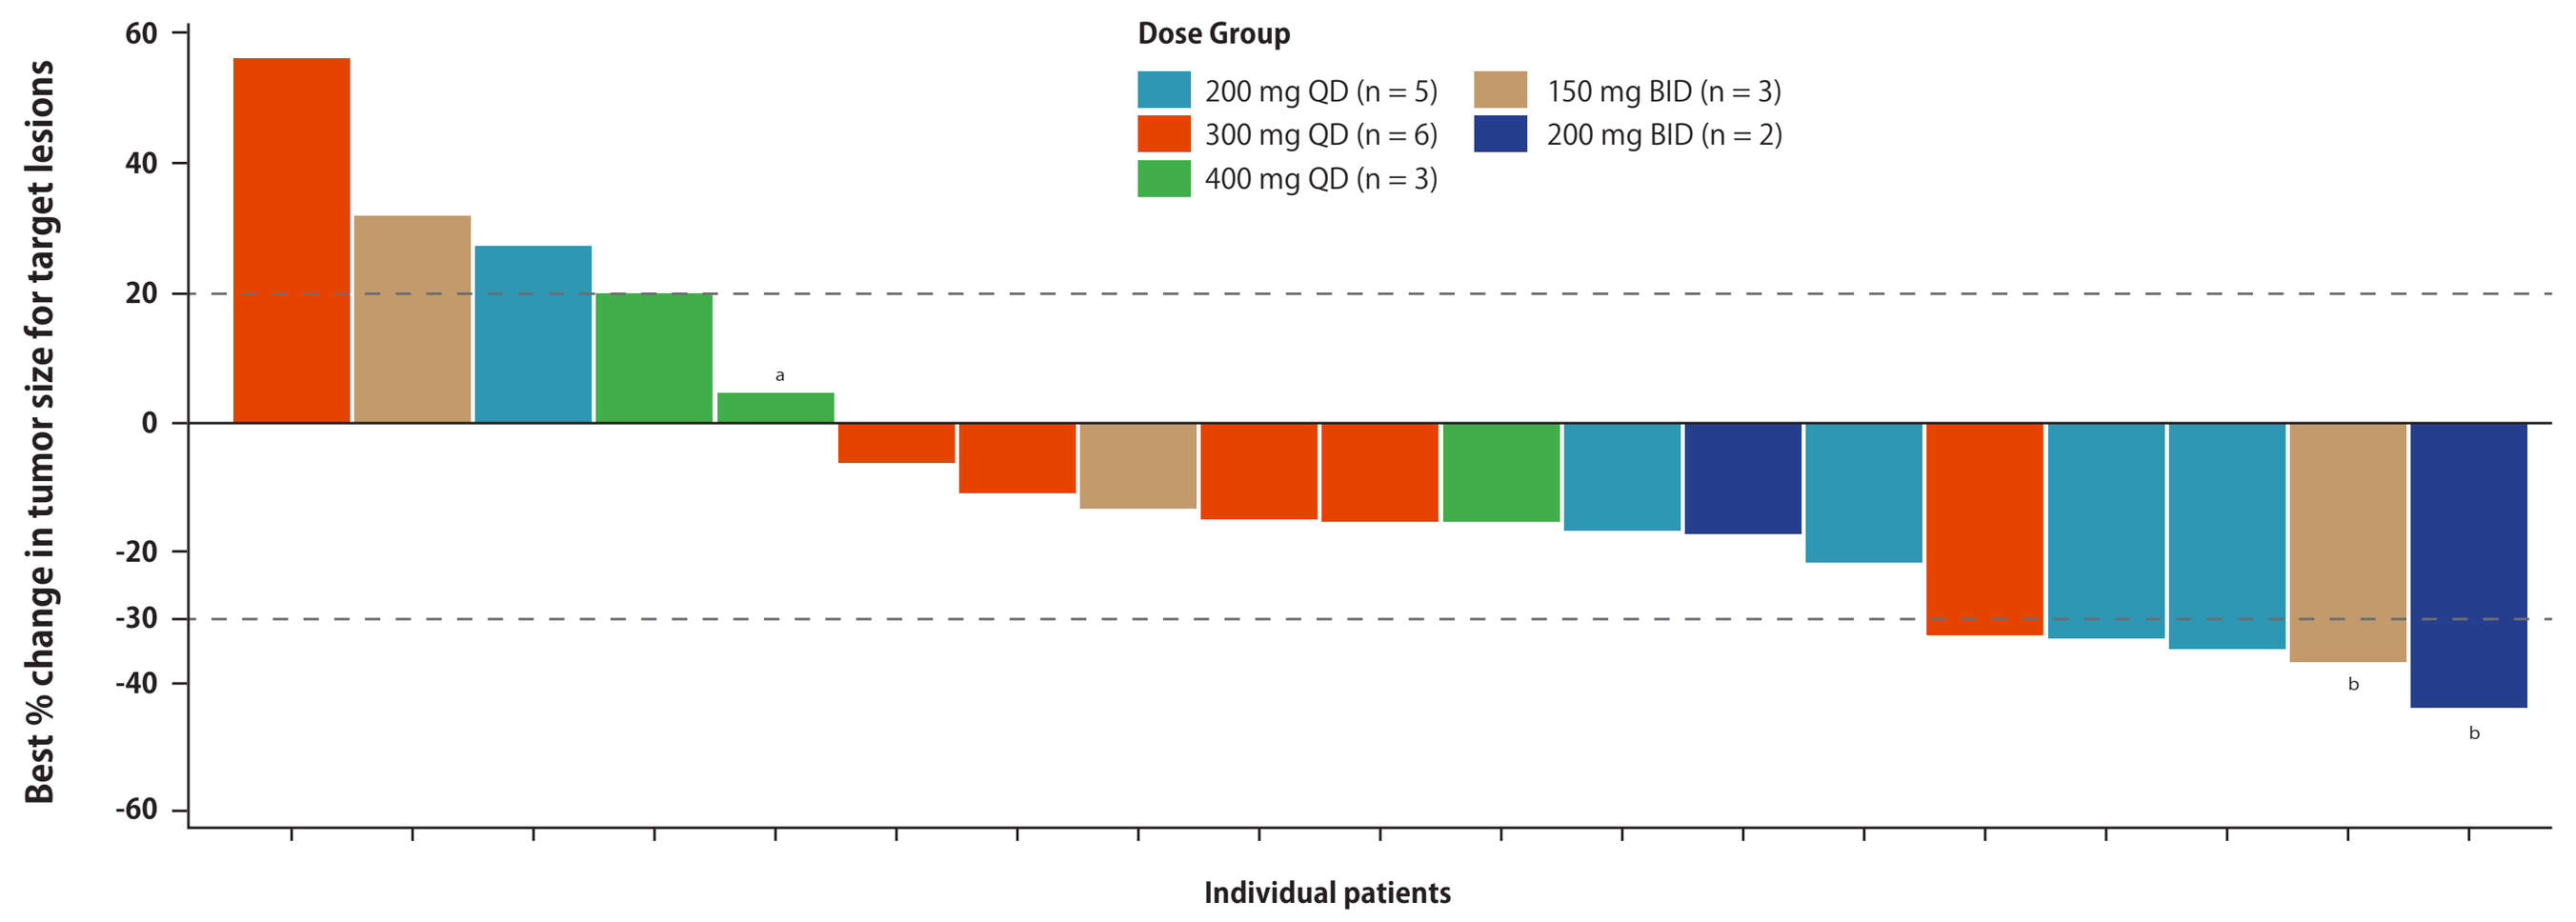
Dose Group
200 mg QD (n = 5)
300 mg QD (n = 6)
400 mg QD (n = 3)
150 mg BID (n = 3)
200 mg BID (n = 2)
Best % change in tumor size for target lesions
Individual patients

Includes all patients who received at least 1 dose of study drug, have measurable target lesions at the baseline, and have at least 1 post baseline tumor assessment. 2 additional patients had clinical progression before their first scheduled post baseline tumor scan. 2 patients had no post baseline tumor measurements, of which 1 patient had disease progression due to a new lesion.
[a] Best overall response is progressive disease due to new lesion; [b] Best overall response is unconfirmed partial response.
BID, twice daily; QD, once daily.

Pharmacokinetics

- PK data are presented in **Table 4**
- Absorption was rapid (median T_{max} ~3 hours), apparent systemic clearance was high (~650 L/h), and the apparent terminal half-life estimated over 48 hours on day 1 has a mean of ~13 hours
- The exposure of a pharmacologically active metabolite, G1T30, was ~10% of parent (lerociclib) exposure
- Exposure increased with dose (C_{max} or $AUC_{0\text{-}inf}$) in an approximately dose-proportional manner
- Coadministration of osimertinib had minimal impact on the PK of lerociclib
 - Overall, C_{max} was similar and $AUC_{0\text{-}inf}$ decreased by ~10% using a linear mixed-effects model compared to lerociclib dosed alone

Table 4. Key Preliminary PK Parameters for Plasma Lerociclib by Dose Group

Lerociclib Dose, mg QD	n	Day −16 (Lerociclib Alone) C_{max}, ng/mL (CV%)	T_{max}, h (min, max)	$AUC_{0\text{-}inf}$, ng*hr/mL (CV%)	$T_{1/2}$, h (CV%)	Day −2 (Lerociclib + Osimertinib) C_{max}, ng/mL (CV%)	T_{max}, h (min, max)	$AUC_{0\text{-}inf}$, ng*hr/mL (CV%)	$T_{1/2}$, h (CV%)
200	7	27.2 (32.8)	3.0 (2.0, 4.0)	325 (33.9)	14.1 (19.2)	17.3 (8.5)	4.0 (3.0, 6.0)	303 (12.2)	14.0 (17.4)
300	3	33.8 (36.6)	3.8 (2.0, 8.0)	395 (23.0)	14.7 (13.0)	26.4 (10.2)	4.0 (3.0, 6.0)	404 (25.3)	12.3 (13.5)
400	4	43.9 (50.0)	6.0 (2.0, 6.0)	710 (48.6)	15.5 (12.9)	42.7 (58.8)	5.0 (4.0, 6.0)	810 (66.2)	12.5 (21.9)

$AUC_{0\text{-}inf}$, area under the curve from time 0 to infinity; C_{max}, maximum observed plasma concentration; CV, coefficient of variation; QD, once daily; T_{max}, time of maximum concentration; $T_{1/2}$, terminal half-life.

Pharmacodynamics

- 16/22 (73%) patients had detectable cfDNA at baseline; 100% of patients with detectable cfDNA had at least 1 EGFR mutation at baseline (**Figure 5**)
- Most common EGFR mutations were Exon 19 deletion and T790M
- 3 patients were sampled at the end of treatment; 1 patient acquired an $EGFR^{C797S}$ mutation, 1 patient acquired a $TP53^{R248Q}$ mutation, and 1 patient didn't acquire any mutations
- Representative plots of cfDNA changes over time are shown in **Figure 6A–F**
- Osimertinib-naïve patients demonstrated a consistent decrease in cfDNA mutant allelic fraction
- Patients who had progressed on prior osimertinib demonstrated more variable results

Figure 5. EGFR Mutation Frequency at Baseline

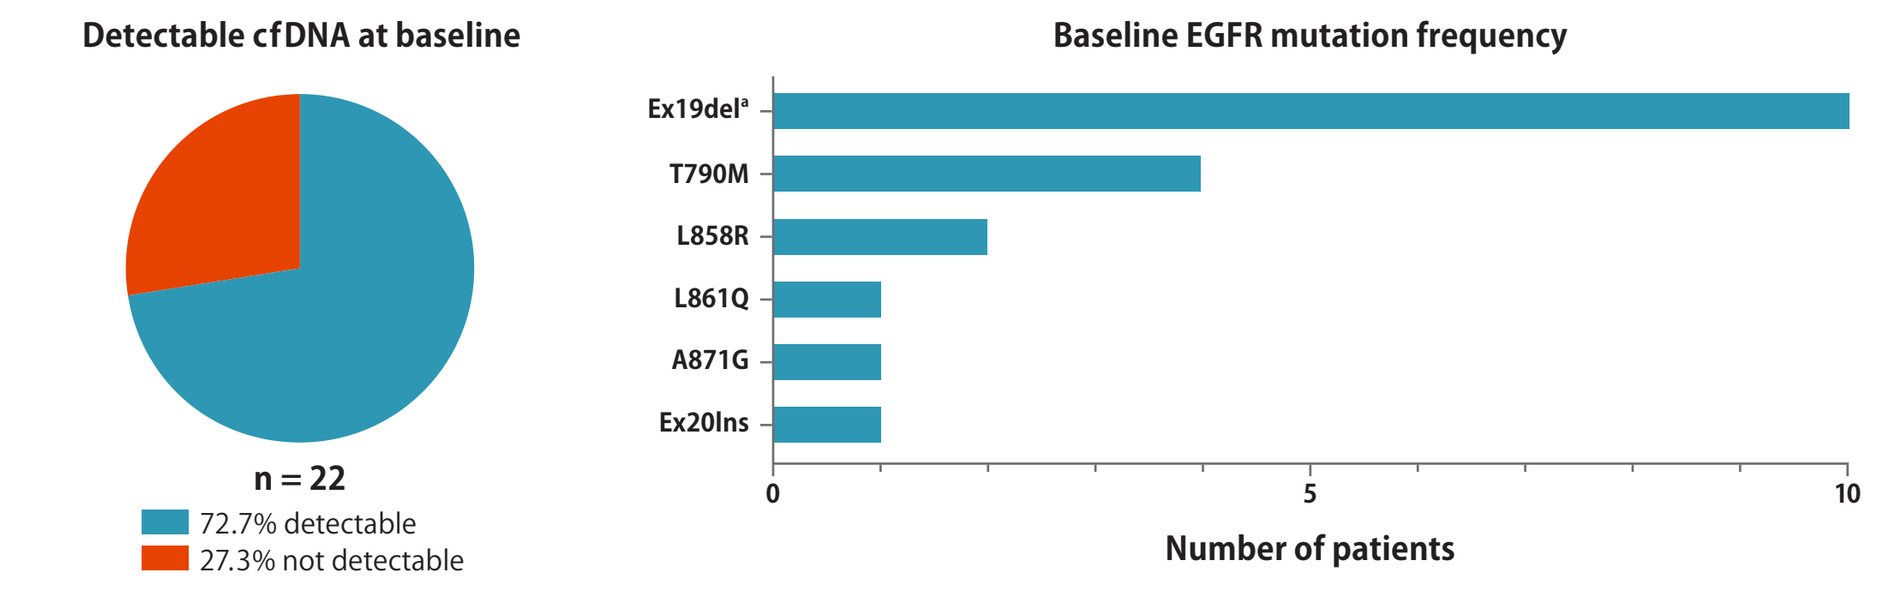
Detectable cfDNA at baseline
n = 22
72.7% detectable
27.3% not detectable
Baseline EGFR mutation frequency
Ex19del[a]
T790M
L858R
L861Q
A871G
Ex20Ins
Number of patients

[a] Ex19del includes: 746-750:ELREA/-, 747-752:LREATS/S, 745-750:KELREA/K, and 746-751:ELREAT/A.
cfDNA, cell-free DNA; EGFR, epidermal growth factor receptor; Ex19del, Exon 19 deletion.

Figure 6. Changes in cfDNA Allele Fraction

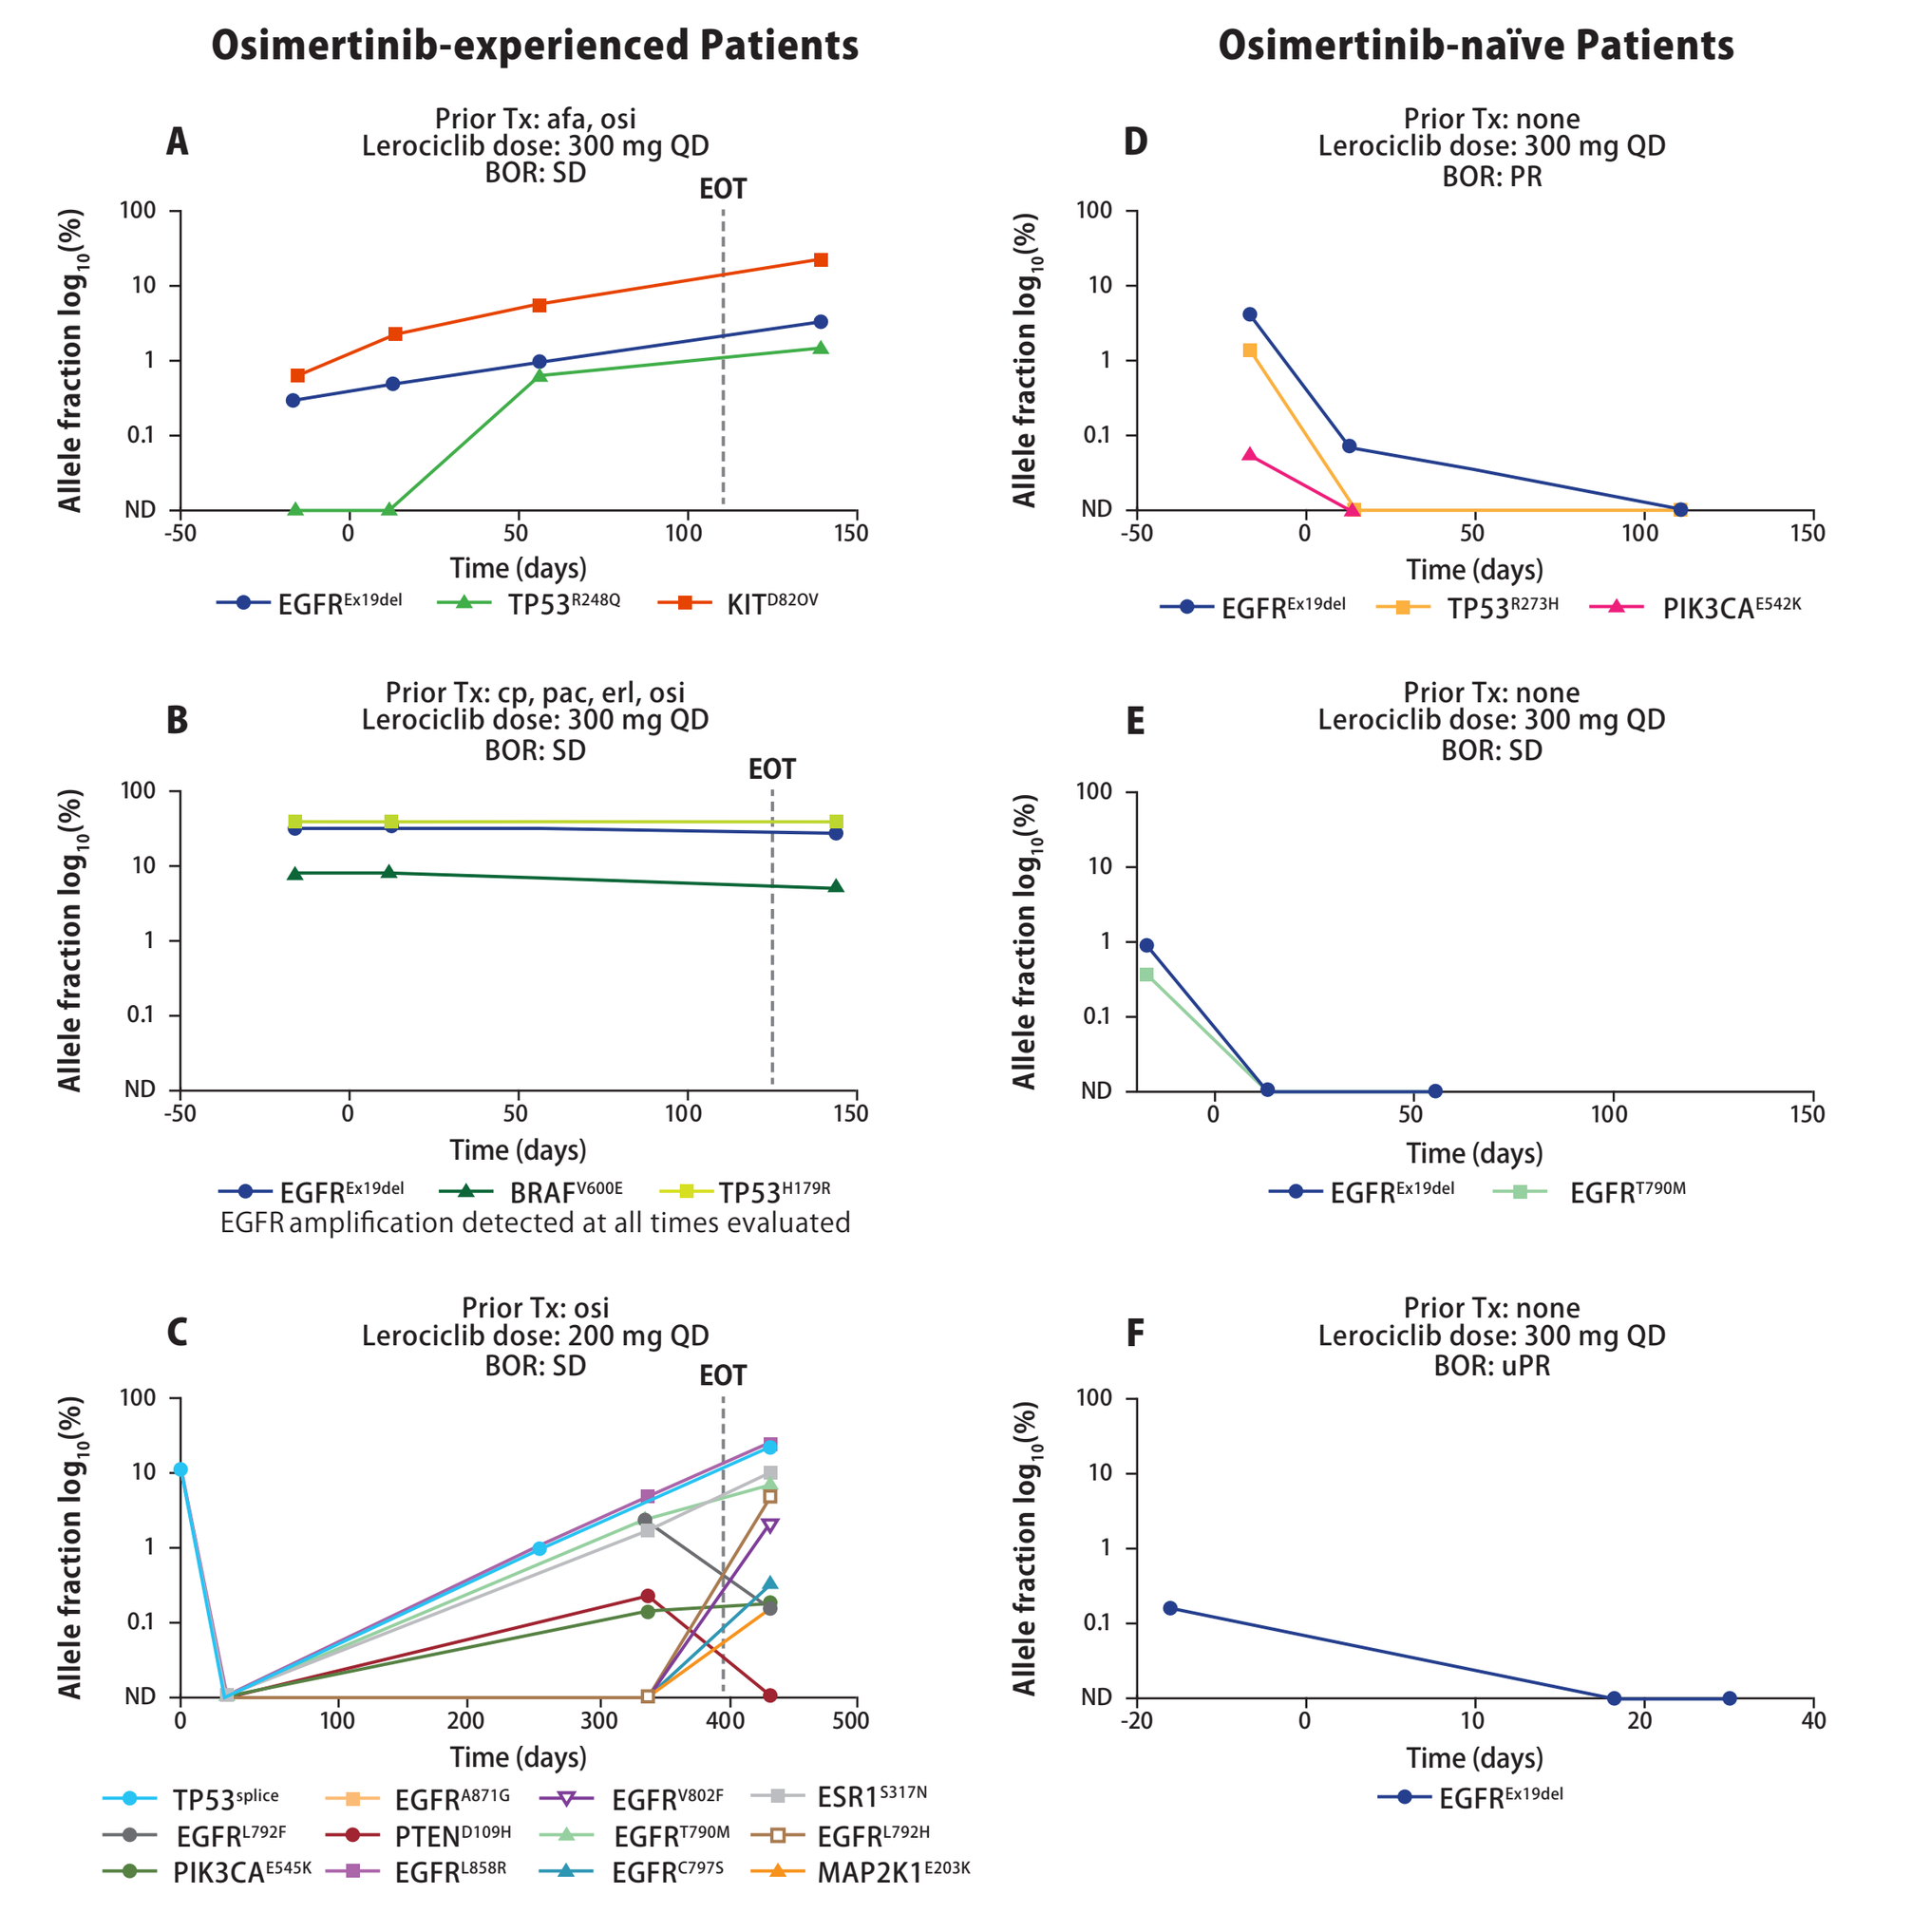
Osimertinib-experienced Patients
Osimertinib-naïve Patients
A
Prior Tx: afa, osi
Lerociclib dose: 300 mg QD
BOR: SD
EOT
EGFR Ex19del
TP53 R248Q
KIT D820V
B
Prior Tx: cp, pac, erl, osi
Lerociclib dose: 300 mg QD
BOR: SD
EGFR Ex19del
BRAF V600E
TP53 H179R
EGFR amplification detected at all times evaluated
C
Prior Tx: osi
Lerociclib dose: 200 mg QD
BOR: SD
TP53 splice
EGFR L792F
PIK3CA E545K
EGFR A871G
PTEN D109H
EGFR L858R
EGFR V802F
EGFR T790M
EGFR C797S
ESR1 S317N
EGFR L792H
MAP2K1 E203K
D
Prior Tx: none
Lerociclib dose: 300 mg QD
BOR: PR
EGFR Ex19del
TP53 R273H
PIK3CA E542K
E
Prior Tx: none
Lerociclib dose: 300 mg QD
BOR: SD
EGFR Ex19del
EGFR T790M
F
Prior Tx: none
Lerociclib dose: 300 mg QD
BOR: uPR
EGFR Ex19del
Allele fraction log10(%)
Time (days)

afa, afatinib; BOR, best overall response; C, cycle; cfDNA, cell-free DNA; cp, carboplatin; D, day; EGFR, epidermal growth factor receptor; EOT, end of treatment; erl, erlotinib; Ex19del, Exon 19 deletion; ND, not detected; osi, osimertinib; pac, paclitaxel; PR, partial response; QD, once daily; SD, stable disease; Tx, treatment; uPR, unconfirmed partial response.

CONCLUSIONS

- The combination of continuously administered lerociclib with osimertinib in patients with EGFR-mutated NSCLC (treatment naïve or previously treated) was well tolerated with preliminary evidence of antitumor activity
- No VTE, QT prolongation or DILI has been reported
- Neutrophil decline plateaued at the end of cycle 1 supporting continuous lerociclib dosing without a drug holiday
- Coadministration of osimertinib had minimal impact on the PK of lerociclib
- MTD for the combination of lerociclib with osimertinib was not defined in this study
- The RP2D is anticipated to be 150 or 200 mg BID

Corresponding author email addresses:
David Berz, DBerz@BHCancerCenter.com; Andrew Beelen, abeelen@g1therapeutics.com

Copies of this poster obtained through QR (Quick Response) and/or text key codes are for personal use only and may not be reproduced without written permission of the authors

ESMO Congress 2019 • September 27 – October 1, 2019 • Barcelona, Spain

ACKNOWLEDGEMENTS
- We thank all the investigators and site staff, with special thanks to the patients and their families
- Study sponsored by G1 Therapeutics. Medical writing assistance was provided by Alligent Europe (Envision Pharma Group), funded by G1 Therapeutics
- We thank Ying Lu and Julio Ruiz for statistical and programming support

CONFLICTS OF INTEREST
DB: honoraria and consulting fees from Boehringer Ingelheim, Genentech, AstraZeneca, Merck, Tempus, and Biocept; major ownership of Valkyrie Therapeutics. **JWG**: research grants and honoraria from AstraZeneca. **YP, CGC, CL, SJ, JAS, RM,** and **APB**: employees of G1 Therapeutics. **AIS, SMG, ICA, JT, TF, SJ**: No conflicts of interest

REFERENCES
1. Takeda M, Nakagawa K. *Int J Mol Sci.* 2019;20:146.
2. Nagano T, et al. *Cells.* 2018;7:212.
3. Bisi JE, et al. *Oncotarget.* 2017;8:42343–58.
4. Bulat I, et al. Poster presentation at ASCO 2018;abstract #1061.
5. Sorrentino JA, et al. Poster presentation at AACR 2018;abstract #1522.
6. Freed DM, et al. Poster presentation at AACR 2019;abstract #4415.