Abstract #339

IMMUNE PROFILING TO INVESTIGATE IMPROVED SURVIVAL IN PATIENTS WITH METASTATIC TRIPLE-NEGATIVE BREAST CANCER RECEIVING TRILACICLIB PRIOR TO CHEMOTHERAPY

Aaron D. Stevens[1]; Joyce O'Shaughnessy[2]; Subing Cao[1]; Jessica Sorrentino[1]; Janet Horton[1]; John S. Yi[1]; and Antoinette R. Tan[3]

[1] G1 Therapeutics, Inc., Research Triangle Park, NC, USA; [2] Baylor University Medical Center, Texas Oncology Dallas, US Oncology Research, Dallas, TX, USA; [3] Levine Cancer Institute, Atrium Health, Charlotte, NC, USA

INTRODUCTION

- Trilaciclib (COSELA™, G1 Therapeutics, Inc.) is an intravenous cyclin-dependent kinase (CDK)4/6 inhibitor indicated to decrease the incidence of chemotherapy-induced myelosuppression in adult patients when administered prior to a platinum/etoposide-containing or topotecan-containing chemotherapy regimen for extensive-stage small cell lung cancer[1]
 - When administered prior to chemotherapy, trilaciclib transiently arrests CDK4/6-dependent hematopoietic stem and progenitor cells and immune cells in the G1 phase of the cell cycle, thus protecting them from chemotherapy-induced damage[1–7]
 - Trilaciclib has also been shown to favorably alter the tumor immune microenvironment through transient T-cell inhibition[2,7–10]
- In a randomized, open-label, phase 2 trial in patients with metastatic triple-negative breast cancer (mTNBC; NCT02978716), administering trilaciclib prior to gemcitabine plus carboplatin (GCb) improved overall survival (OS; secondary endpoint) compared with GCb alone (median 19.8 vs 12.6 months; *P* < 0.0001)[9,10]
- Subgroup analyses suggested that:
 - Administering trilaciclib prior to GCb prolonged OS irrespective of programmed death-ligand 1 (PD-L1) status but had greater benefit in the PD-L1–positive population (**Table**)[10]
 - Survival benefits with trilaciclib were more pronounced in, but not exclusive to, patients with higher immune-related gene expression[10]
 - Administering trilaciclib resulted in an enrichment of new T-cell clones and decreased Simpson clonality in peripheral blood, suggesting enhanced T-cell activation[10]
- The current research aimed to further investigate potential immune mechanisms of antitumor efficacy among patients receiving trilaciclib prior to GCb

Table. Subgroup Analysis of OS According to PD-L1 Status[10]

	PD-L1 Positive		PD-L1 Negative	
	GCb Alone	Trilaciclib Prior to GCb	GCb Alone	Trilaciclib Prior to GCb
Patients, n	17	32	10	26
Median OS, months (95% CI)	10.5 (6.3–18.8)	32.7 (17.7–NR)	13.9 (12.6–NR)	17.8 (13.1–NR)
HR (95% CI)		0.34 (0.2–0.7)		0.48 (0.2–1.2)

GCb, gemcitabine plus carboplatin; HR, hazard ratio; NR, not reached; OS, overall survival; PD-L1, programmed death-ligand 1.

METHODS

Study Design and Participants

- Full details of the clinical trial design have been published previously[9,10]
 - Briefly, patients were randomized (1:1:1) to: group 1 (GCb alone on days 1 and 8), group 2 (trilaciclib prior to GCb on days 1 and 8), and group 3 (trilaciclib alone on days 1 and 8, and trilaciclib before GCb on days 2 and 9)
 - Prespecified secondary antitumor endpoints included objective response rate, progression-free survival (both per Response Evaluation Criteria in Solid Tumours [RECIST] v1.1), and OS
- Here, data were analyzed following a data cut-off of May 15, 2020 (for response) and the final database lock on July 17, 2020 (for OS). Trilaciclib-treated patients (groups 2 and 3) were combined into a single cohort for further analysis based on similar OS improvement and data readouts
 - Patients were defined as responders (confirmed complete or partial response) or nonresponders (stable or progressive disease) according to RECIST v1.1

Analysis of Immune-Cell Subsets and Activation Markers in Blood and Tumors

- PD-L1 expression was assessed in diagnostic tumor tissue samples from each patient using the Ventana SP142 PD-L1 assay (Ventana Medical Systems, Inc., Tuscon, AZ, USA)[11]
 - PD-L1 expression was scored as negative or positive if < 1% or ≥ 1% of the total tumor area contained PD-L1–labeled immune cells, respectively[11]
- Peripheral blood was collected prior to and during treatment for flow cytometric analysis
- Genomic DNA and total RNA were simultaneously purified from formalin-fixed, paraffin-embedded, diagnostic tumor samples using the AllPrep DNA/RNA FFPE kit (QIAGEN, Germantown, MD, USA), and libraries were prepared using TruSeq RNA and DNA Exome kits for RNA-Seq and DNA-Seq, respectively (Illumina, San Diego, CA, USA)
 - Cluster generation and sequencing of libraries was performed on the Illumina HiSeq system, and gene expression read counts and fragments per kilobase of exon per million mapped reads (FPKM) were quantified using RNA-Seq by Expectation Maximization (RSEM) software[12]
- Differential gene expression analysis between responders and nonresponders was performed using the DESeq2 package,[13] and related pathways identified by Gene Set Enrichment Analysis (GSEA) software[14,15] using the Kyoto Encyclopedia of Genes and Genomes (KEGG) database
- Tumor inflammation signatures[16] were used to assess the tumor immune microenvironment

RESULTS

- Of 68 patients who received trilaciclib prior to GCb, antitumor response status was available for 58, comprising 27 responders (46.6%) and 31 nonresponders (53.4%)
 - PD-L1 status was available for 22 of 27 responders and 27 of 31 nonresponders
 - Among responders, 15 were PD-L1 positive and 7 were PD-L1 negative
 - Among nonresponders, 15 were PD-L1 positive and 12 were PD-L1 negative
- After 2 cycles, patients who received trilaciclib prior to GCb had fewer, but more functional T cells and fewer myeloid-derived suppressor cells (MDSCs) than patients who received GCb alone (**Figure 1**)

Figure 1. Changes to (A) Immune-Cell Populations and (B, C) T-cell Function in Peripheral Blood Over 2 Cycles (C1D1 vs C3D1) for Trilaciclib Plus GCb Versus GCb Alone

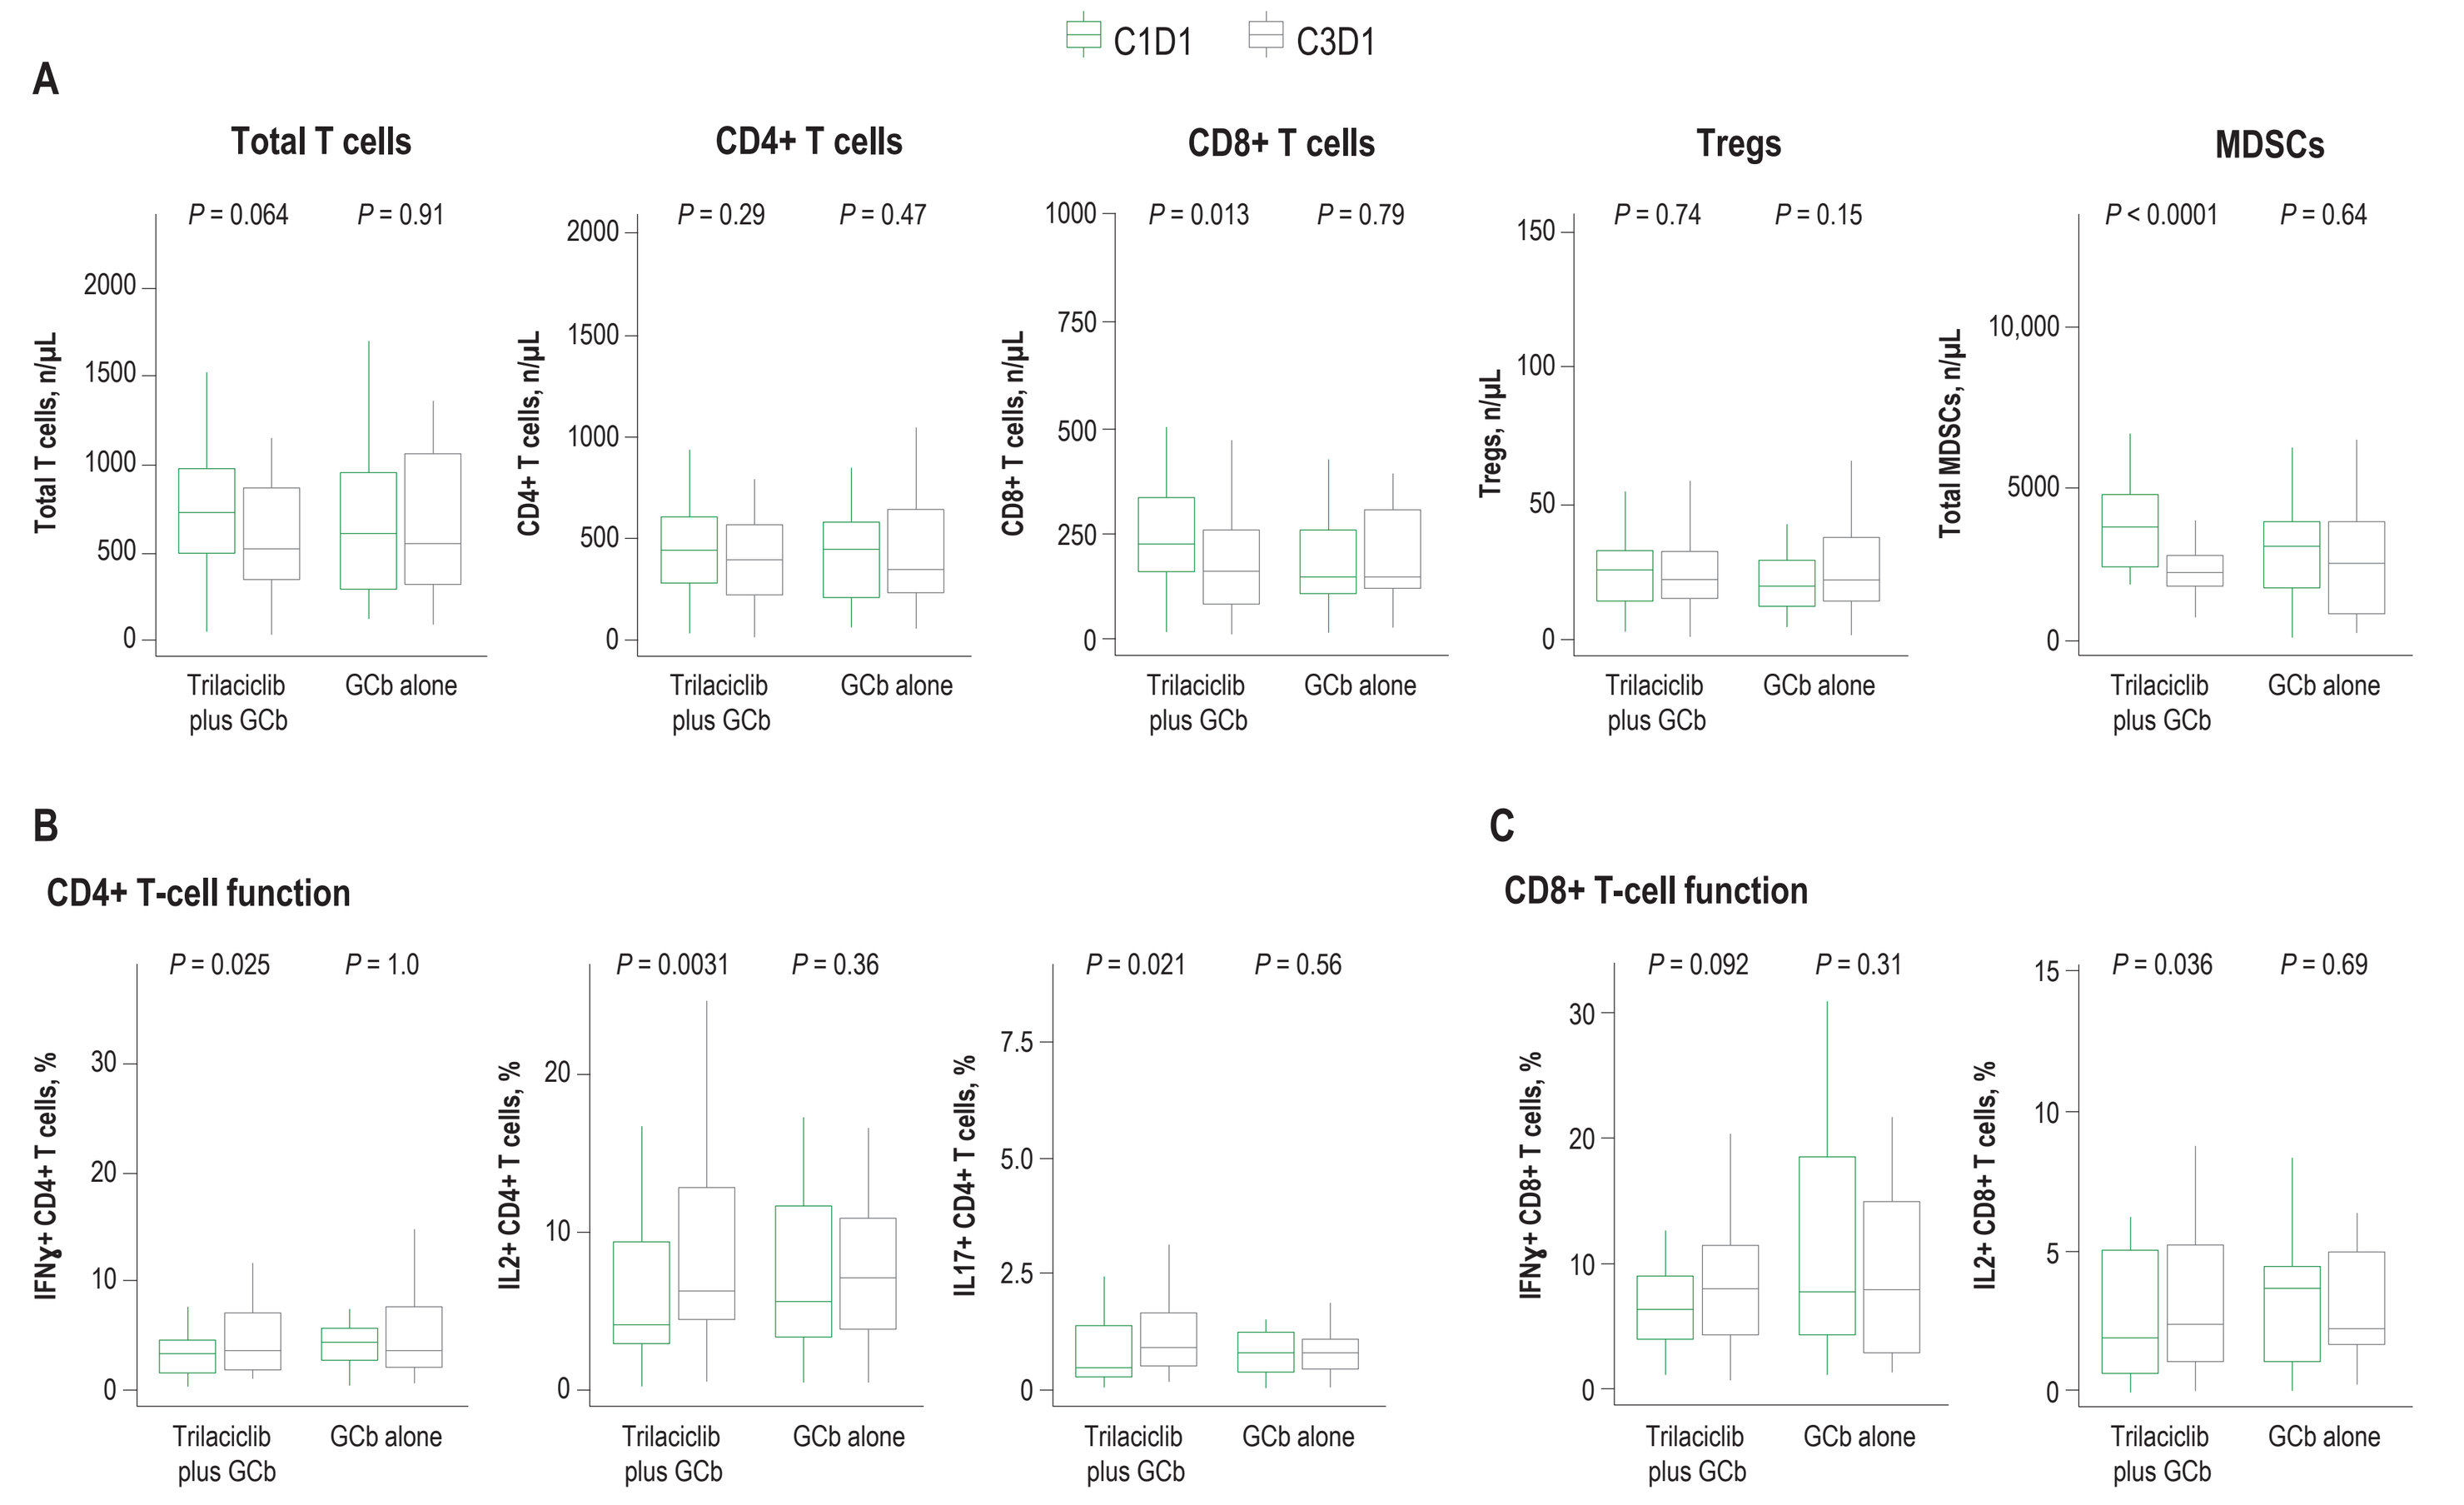
C1D1
C3D1
A
Total T cells
P = 0.064
P = 0.91
CD4+ T cells
P = 0.29
P = 0.47
CD8+ T cells
P = 0.013
P = 0.79
Tregs
P = 0.74
P = 0.15
MDSCs
P < 0.0001
P = 0.64
Total T cells, n/μL
CD4+ T cells, n/μL
CD8+ T cells, n/μL
Tregs, n/μL
Total MDSCs, n/μL
Trilaciclib plus GCb
GCb alone
B
CD4+ T-cell function
P = 0.025
P = 1.0
P = 0.0031
P = 0.36
P = 0.021
P = 0.56
IFNγ+ CD4+ T cells, %
IL2+ CD4+ T cells, %
IL17+ CD4+ T cells, %
C
CD8+ T-cell function
P = 0.092
P = 0.31
P = 0.036
P = 0.69
IFNγ+ CD8+ T cells, %
IL2+ CD8+ T cells, %

C, cycle; D, day; GCb, gemcitabine plus carboplatin; IFNγ, interferon gamma; IL, interleukin; MDSC, myeloid-derived suppressor cell; Treg, regulatory T cell.

- Analysis of tumor samples with RNA-Seq data revealed 69 differentially expressed genes between trilaciclib responders (n = 15) and nonresponders (n = 17) (**Figure 2A**)
 - KEGG pathways upregulated in trilaciclib responders included T-cell receptor signaling, antigen processing and presentation, natural killer cell–mediated cytotoxicity, NOD-like receptor signaling, Toll-like receptor signaling, cytosolic DNA sensing, graft-versus-host disease, and glycosphingolipid biosynthesis
 - Analysis of immune gene signatures revealed a higher T-cell exhaustion score at baseline among responders versus nonresponders (*P* = 0.054; **Figure 2B**)

Figure 2. (A) Differential Gene Expression Analysis and (B) T-cell Exhaustion in Tumor Samples from Trilaciclib Responders and Nonresponders

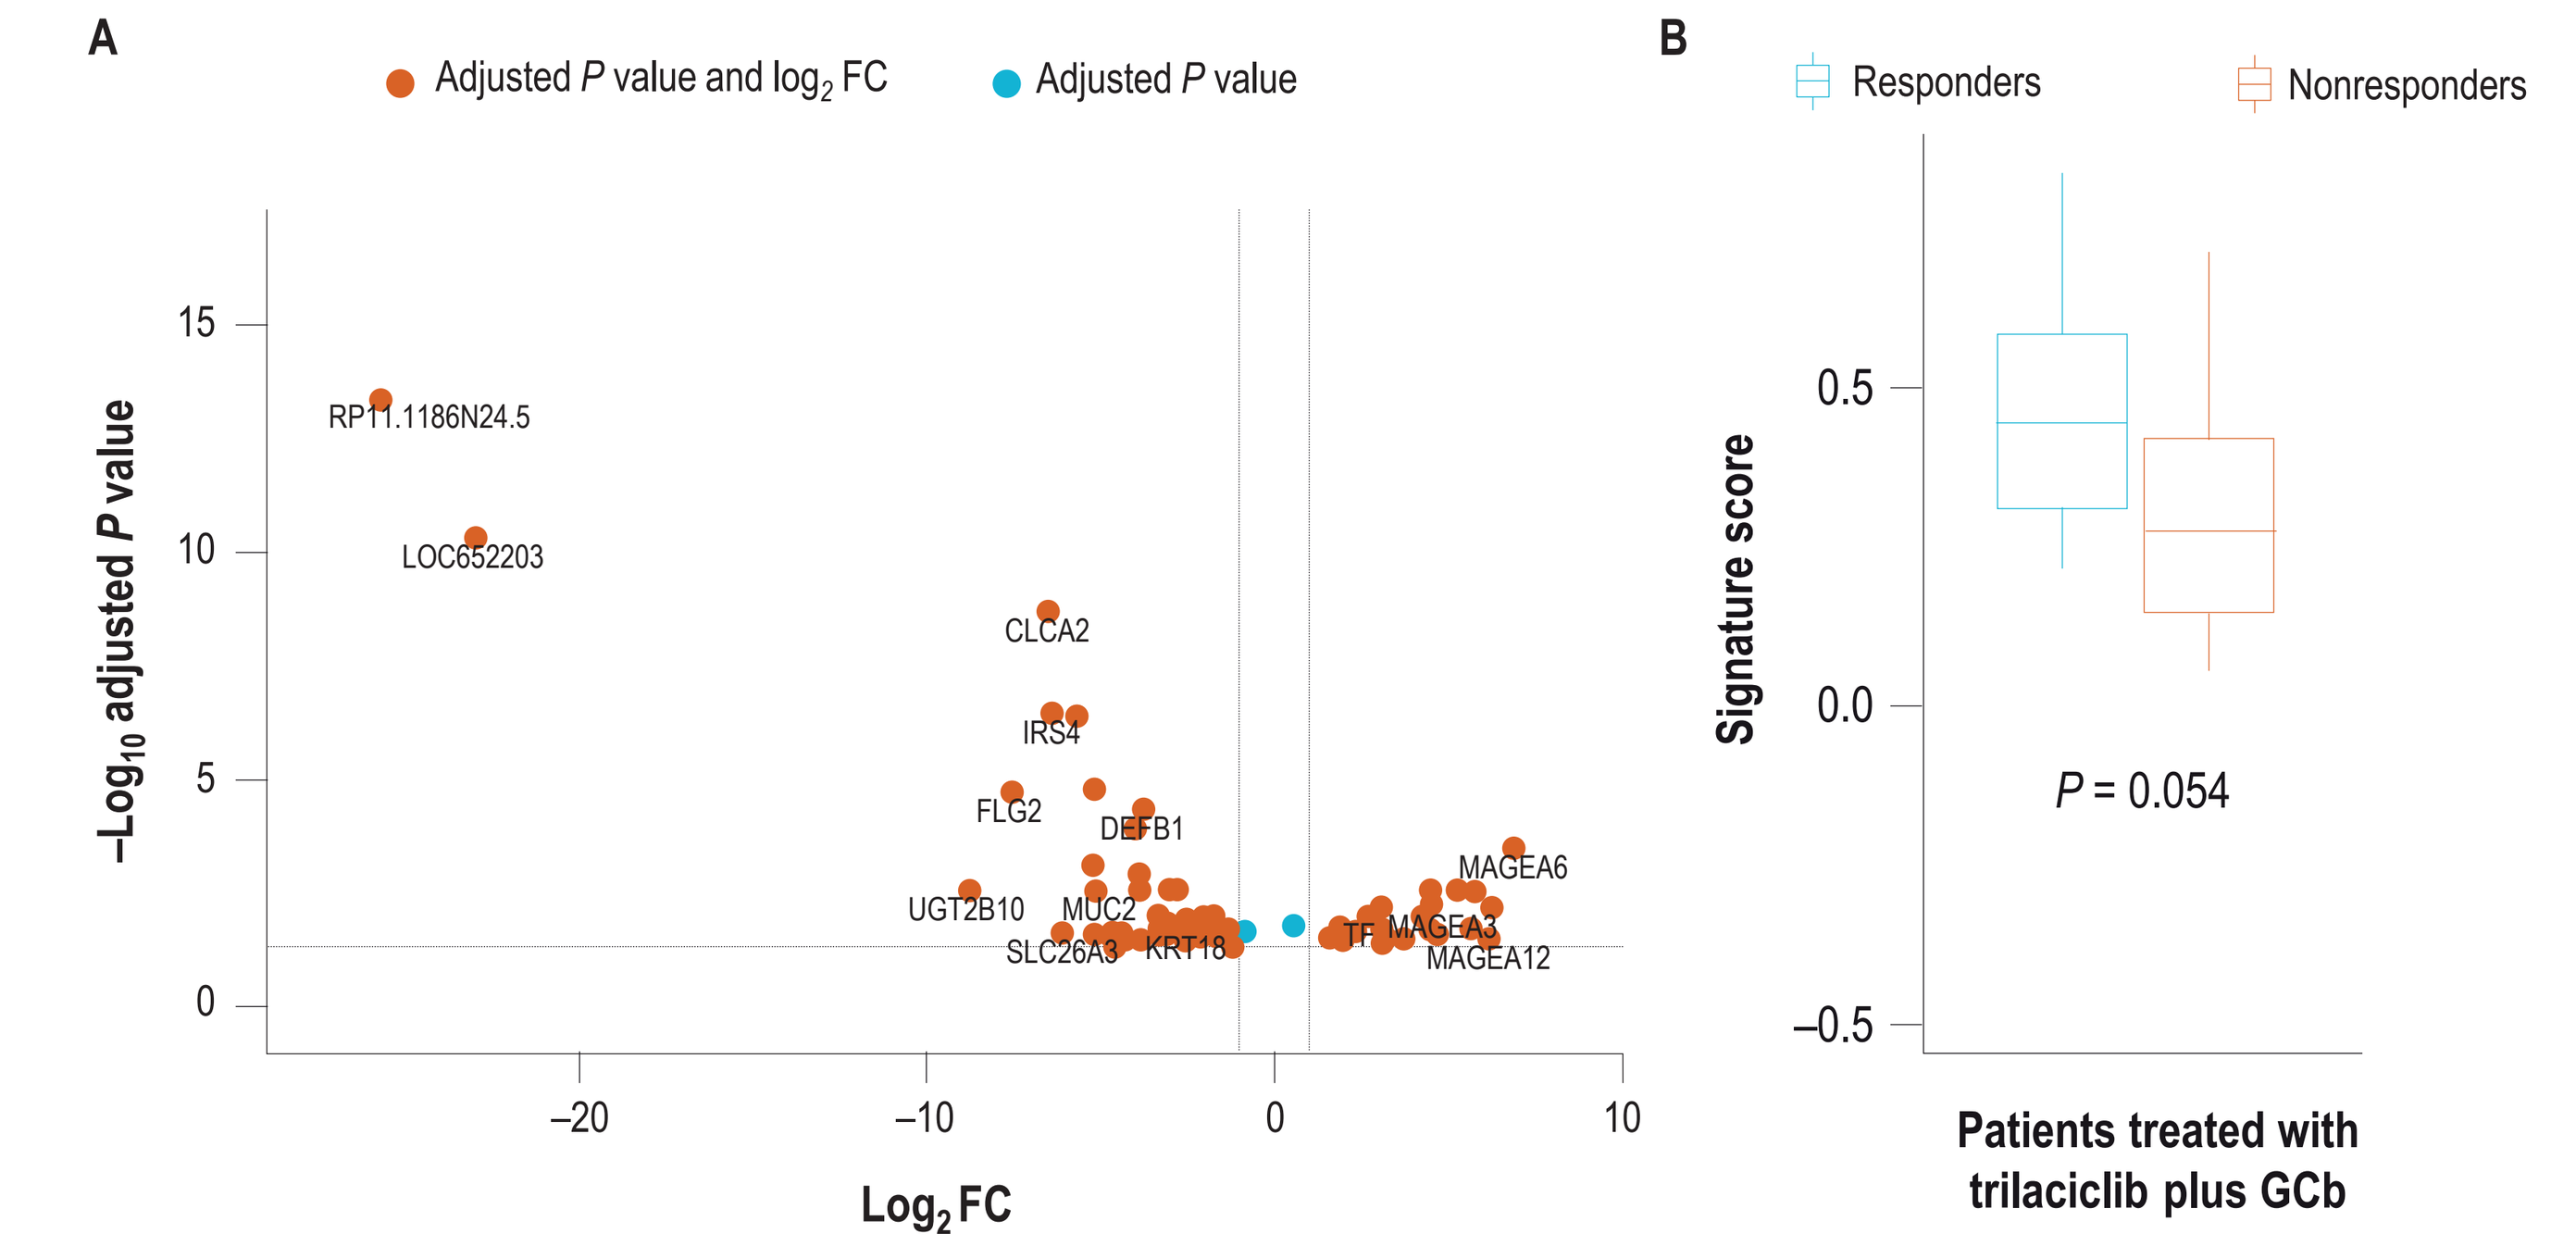
A
Adjusted P value and log2 FC
Adjusted P value
–Log10 adjusted P value
Log2 FC
RP11.1186N24.5
LOC652203
CLCA2
IRS4
FLG2
DEFB1
UGT2B10
MUC2
SLC26A3
KRT18
MAGEA6
MAGEA3
MAGEA12
B
Responders
Nonresponders
Signature score
P = 0.054
Patients treated with trilaciclib plus GCb

Patients with an antitumor response following treatment with trilaciclib plus GCb had 69 differentially expressed genes versus nonresponders at a false discovery rate of < 0.05; 23 genes were upregulated, and 46 genes were downregulated.
FC, fold change; GCb, gemcitabine plus carboplatin.

- After 2 cycles:
 - T-cell numbers were maintained in patients with an antitumor response to trilaciclib plus GCb but reduced in patients without a response; both responders and nonresponders had reduced MDSCs (**Figure 3**)
 - T-cell function was maintained or improved in responders versus maintained or reduced in nonresponders (**Figure 4**)
 - Human leukocyte antigen – DR isotype expression, a marker of T-cell activation, was also downregulated in nonresponders (**Figure 4**)

Figure 3. Changes to Immune-Cell Populations in Peripheral Blood Over 2 Cycles (C1D1 vs C3D1) for Trilaciclib Responders Versus Nonresponders

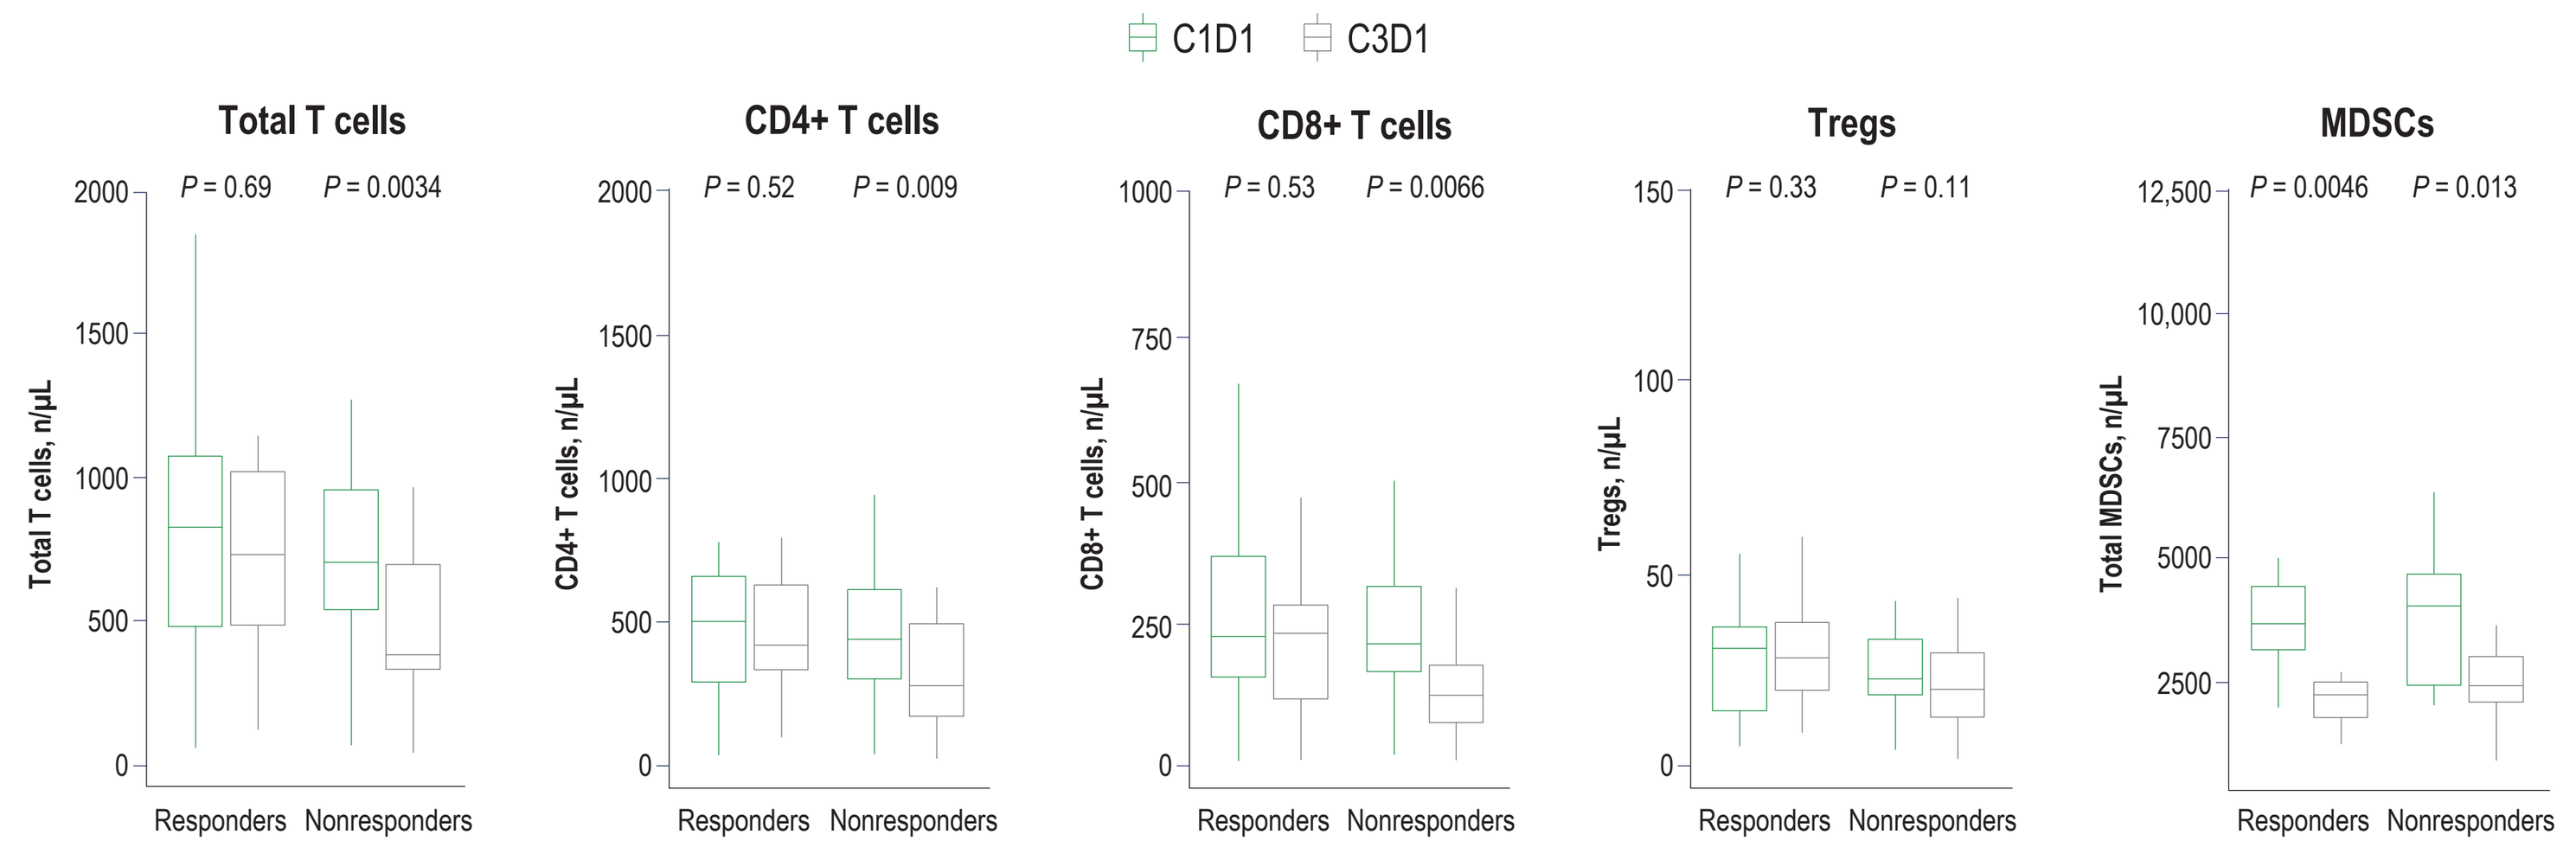
C1D1
C3D1
Total T cells
P = 0.69
P = 0.0034
CD4+ T cells
P = 0.52
P = 0.009
CD8+ T cells
P = 0.53
P = 0.0066
Tregs
P = 0.33
P = 0.11
MDSCs
P = 0.0046
P = 0.013
Total T cells, n/μL
CD4+ T cells, n/μL
CD8+ T cells, n/μL
Tregs, n/μL
Total MDSCs, n/μL
Responders
Nonresponders

C, cycle; D, day; MDSC, myeloid-derived suppressor cell; Treg, regulatory T cell.

Figure 4. Changes to (A) CD4+ and (B) CD8+ T-cell Function in Peripheral Blood Over 2 Cycles (C1D1 vs C3D1) for Trilaciclib Responders Versus Nonresponders

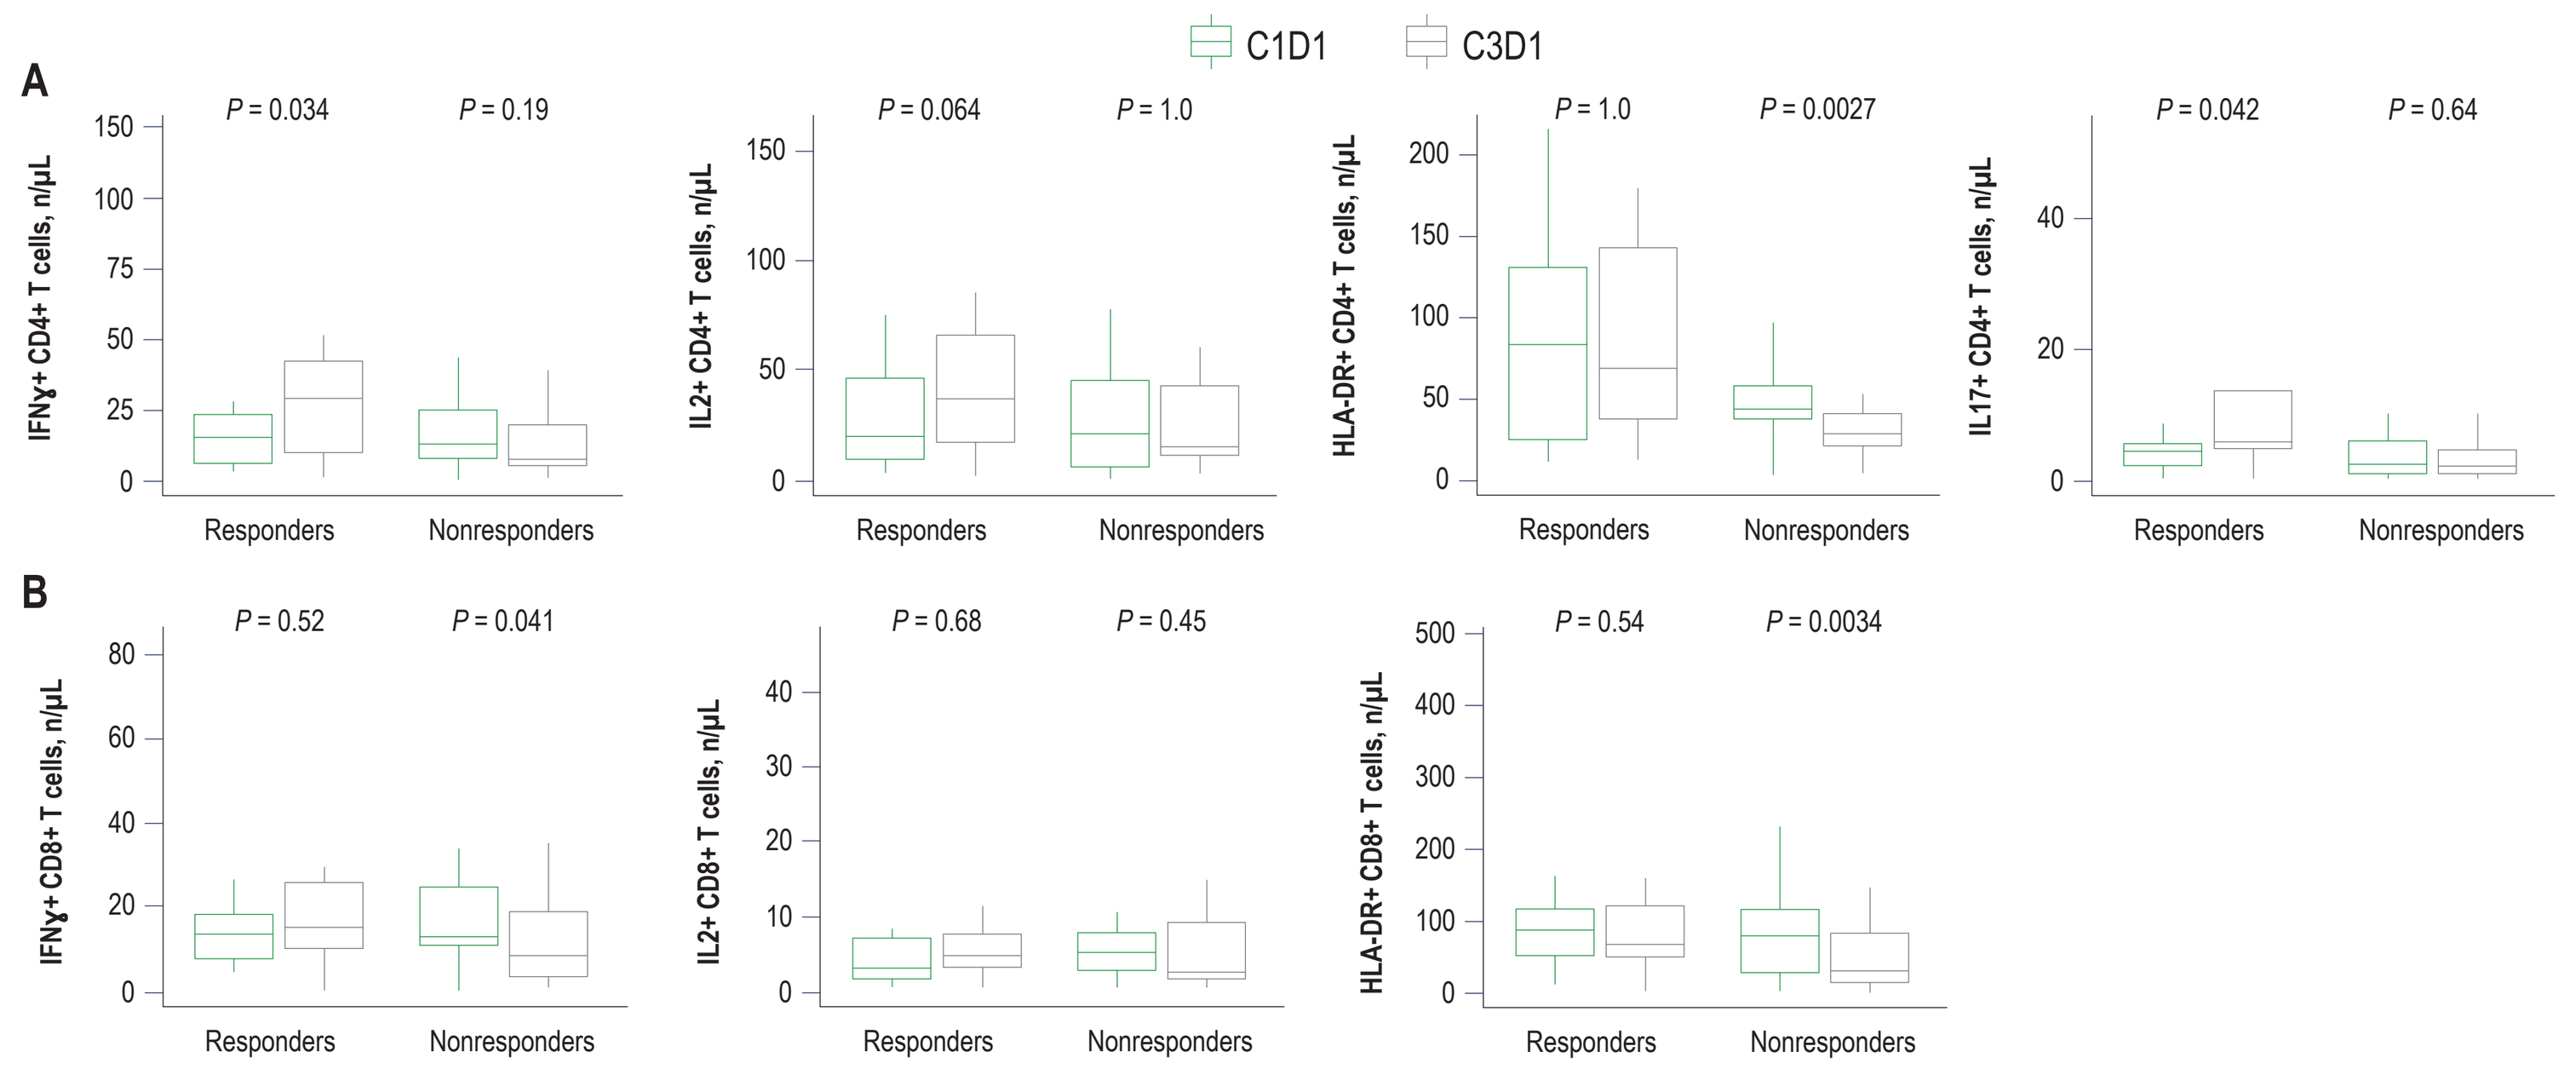
C1D1
C3D1
A
P = 0.034
P = 0.19
P = 0.064
P = 1.0
P = 1.0
P = 0.0027
P = 0.042
P = 0.64
IFNγ+ CD4+ T cells, n/μL
IL2+ CD4+ T cells, n/μL
HLA-DR+ CD4+ T cells, n/μL
IL17+ CD4+ T cells, n/μL
B
P = 0.52
P = 0.041
P = 0.68
P = 0.45
P = 0.54
P = 0.0034
IFNγ+ CD8+ T cells, n/μL
IL2+ CD8+ T cells, n/μL
HLA-DR+ CD8+ T cells, n/μL
Responders
Nonresponders

C, cycle; D, day; HLA-DR, human leukocyte antigen – DR isotype; IFNγ, interferon gamma; IL, interleukin.

- Among trilaciclib-treated patients with PD-L1–positive tumors, responders had a trend toward an enriched tumor inflammation signature score compared with nonresponders (**Figure 5**)
- Responders with both PD-L1–positive and –negative tumors had increased numbers of memory CD8 T cells and naïve CD8 T cells after 2 cycles compared with nonresponders (data not shown)

Figure 5. TIS in Tumor Samples from Trilaciclib Responders Versus Nonresponders According to PD-L1 Status

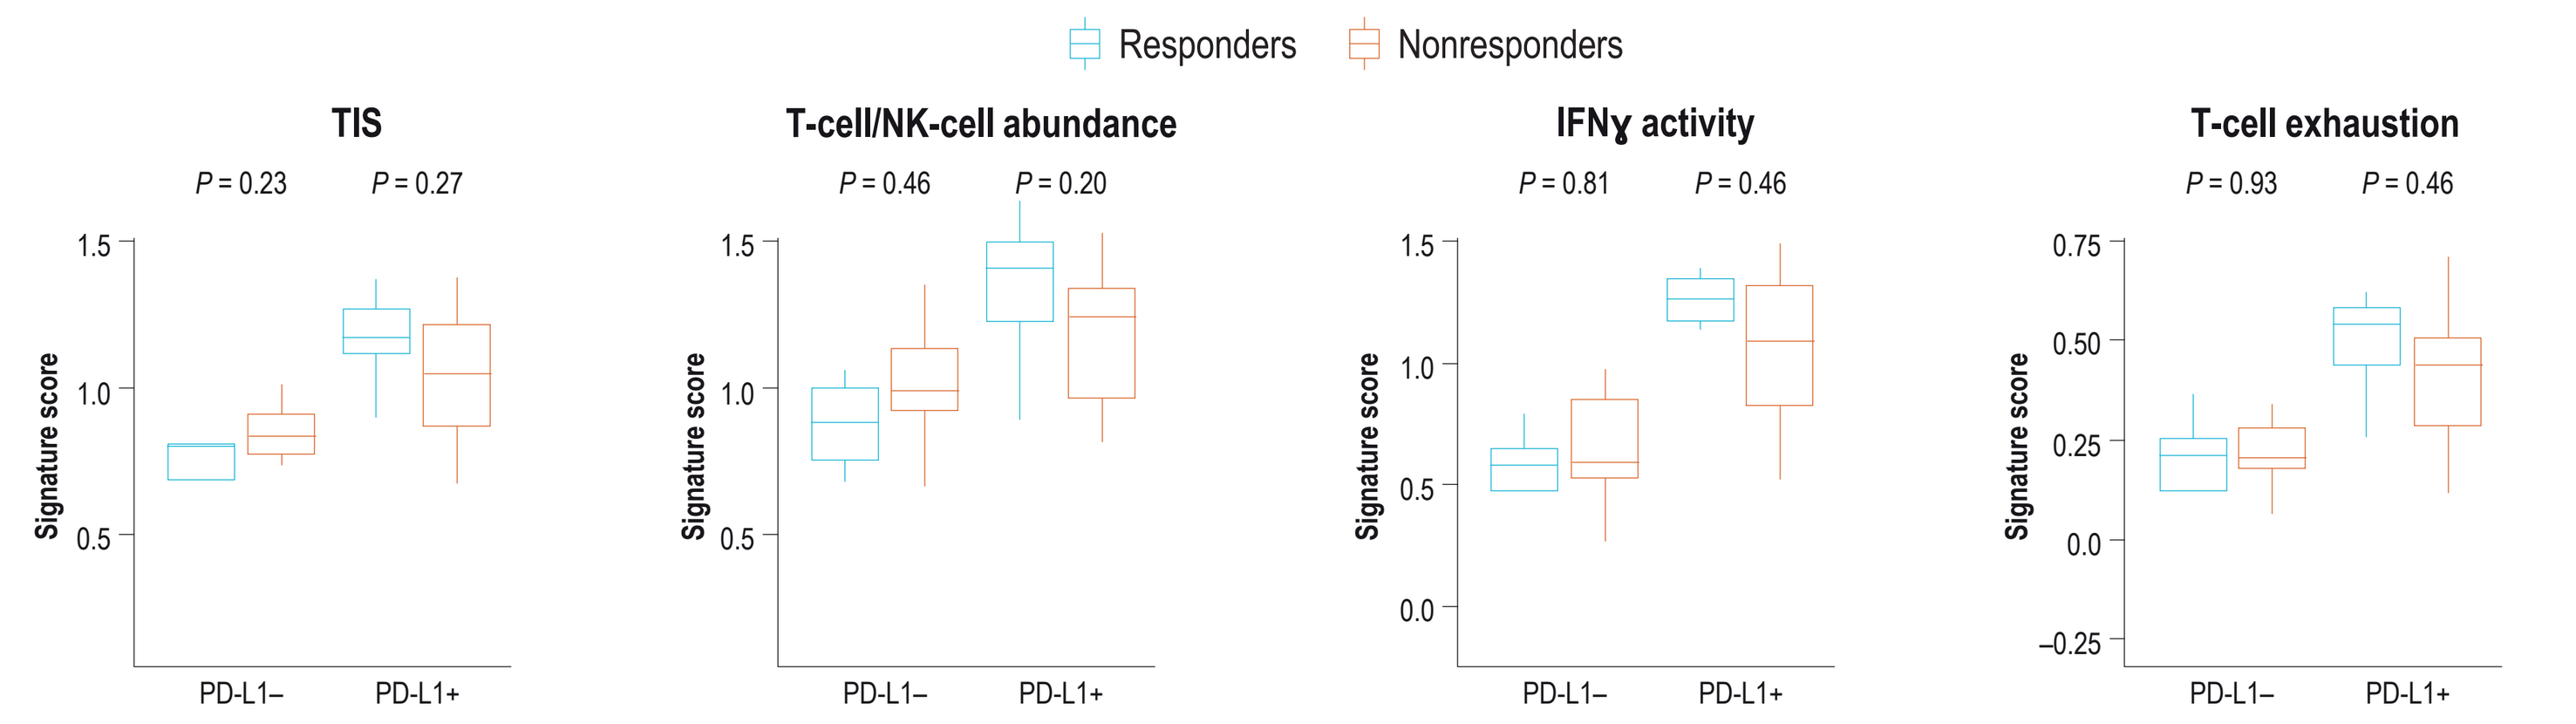
Responders
Nonresponders
TIS
P = 0.23
P = 0.27
T-cell/NK-cell abundance
P = 0.46
P = 0.20
IFNγ activity
P = 0.81
P = 0.46
T-cell exhaustion
P = 0.93
P = 0.46
Signature score
PD-L1–
PD-L1+

TIS is an investigational 18-gene signature that detects an adaptive immune response within tumors by measuring expression of genes associated with antigen presentation, T-cell/NK-cell abundance, IFNγ activity, and T-cell exhaustion.[16]
IFNγ, interferon gamma; NK, natural killer; PD-L1, programmed death-ligand 1; PD-L1–, PD-L1 negative; PD-L1+, PD-L1 positive; TIS, tumor inflammation signature.

CONCLUSIONS

- The data suggest that administering trilaciclib prior to GCb may enhance antitumor efficacy by modulating the composition and response of immune-cell subsets
- Compared with nonresponders, responders had an upregulation of genes involved in immune-system activation and had higher T-cell exhaustion scores at baseline, potentially reflecting more T-cell infiltration and a greater existing immune response
- Although patients receiving trilaciclib had fewer peripheral T cells after 2 cycles, those T cells were more functional, as evidenced by an increase in the number of cytokine-producing cells
- Greater peripheral immune responses were observed at baseline among PD-L1–positive responders versus nonresponders
- The impact of trilaciclib on changes to the tumor-infiltrating immune response will be further investigated in the phase 3 PRESERVE 2 trial in patients with mTNBC (NCT04799249) and in a planned mechanism-of-action trial in the neoadjuvant TNBC setting

Society for Immunotherapy of Cancer 36th Annual Meeting
November 10–14, 2021 | Washington, DC, USA

REFERENCES:
1. COSELA™ (trilaciclib). Prescribing Information. https://www.g1therapeutics.com/cosela/pi/. Accessed September 2021.
2. Daniel D, et al. *Int J Cancer*. 2021;148:2557–70.
3. Weiss JM, et al. *Ann Oncol*. 2019;30:1613–21.
4. Hart LL, et al. *Adv Ther*. 2021;38:350–65.
5. He S, et al. *Sci Transl Med*. 2017;9:eaal3986.
6. Li C, et al. *Cancer Chemother Pharmacol*. 2021;87:689–700.
7. Lai A, et al. *J Immunother Cancer*. 2020;8:e000847.
8. Deng J, et al. *Cancer Discov*. 2018;8:216–33.
9. Tan AR, et al. *Lancet Oncol*. 2019;20:1587–601.
10. O'Shaughnessy J, et al. SABCS 2020 poster presentation: poster #PD1-06.
11. US Food and Drug Administration. VENTANA PD-L1 (SP142) Assay. https://www.accessdata.fda.gov/cdrh_docs/pdf16/p160002s009c.pdf. Accessed September 2021.
12. Li B, Dewey CN. *BMC Bioinformatics*. 2011;12:323.
13. Love MI, et al. *Genome Biol*. 2014;15:550.
14. Subramanian A, et al. *Proc Natl Acad Sci USA*. 2005;102:15545–50.
15. Mootha VK, et al. *Nat Genet*. 2003;34:267–73.
16. Danaher P, et al. *J Immunother Cancer*. 2018;6:63.

ACKNOWLEDGMENTS:
- Study sponsored by G1 Therapeutics, Inc.
- Flow cytometry and RNA sequencing analyses were performed by Covance, Inc., and Q2 Laboratory Solutions, respectively
- Medical writing assistance was provided by Alligent Europe (Envision Pharma Group), funded by G1 Therapeutics, Inc.