384

MYELOPRESERVATION AND REDUCED USE OF SUPPORTIVE CARE WITH TRILACICLIB IN PATIENTS WITH SMALL CELL LUNG CANCER

Jared Weiss[1]; Jerome Goldschmidt[2]; Zoran Andric[3]; Konstantin H. Dragnev[4]; Yili Pritchett[5]; Shannon R. Morris[5]; Rajesh K. Malik[5]; and Davey B. Daniel[6]

[1] Lineberger Comprehensive Cancer Center, University of North Carolina, Chapel Hill, NC, USA; [2] US Oncology Research, McKesson Specialty Health, Blacksburg, VA, USA; [3] University Hospital Medical Center Bezanijska Kosa, Belgrade, Serbia; [4] Norris Cotton Cancer Center Dartmouth-Hitchcock, Lebanon, NH, USA; [5] G1 Therapeutics, Inc., Research Triangle Park, NC, USA; [6] Sarah Cannon Research Institute, Nashville, TN, USA

BACKGROUND

- Chemotherapy (CT)-induced myelosuppression (CIM) can negatively impact patients' quality of life, and their ability to receive CT on time and at standard-of-care doses
 - Current CIM interventions are specific to individual hematopoietic cell lineages, reactively administered, and impart their own set of risks for side effects and adverse reactions[1]
- Trilaciclib is an intravenous (IV) CDK4/6 inhibitor that is administered prior to CT to reduce the risk of CIM
 - Trilaciclib transiently arrests normal cells in the G_1 phase of the cell cycle during CT exposure to preserve bone marrow and immune system function from CT-induced damage (myelopreservation)
- The effects of trilaciclib have been investigated in 3 randomized, double-blind, placebo-controlled, phase 2 studies in adult patients with extensive-stage small cell lung cancer (ES-SCLC)[1–3]
- The aim of this analysis was to pool data from these studies to understand the myelopreservation effects of trilaciclib with greater statistical precision

METHODS

Study Design

- Data were pooled from patients enrolled in the studies outlined in **Table 1**
 - In each study, patients received IV trilaciclib 240 mg/m^2 or placebo on each day prior to CT administration

Table 1. Overview of Trilaciclib Clinical Trials Included in Pooled Analysis

Study	Patient population	Treatment schedule
G1T28-02 (NCT02499770)	1L ES-SCLC	Trilaciclib or placebo on days 1–3 of each 21-day E/P cycle[a]
G1T28-05 (NCT03041311)	1L ES-SCLC	Trilaciclib or placebo on days 1–3 of each 21-day E/P/A cycle for up to 4 cycles (induction), followed by A every 21 days (maintenance)[b]
G1T28-03 (NCT02514447)	2/3L ES-SCLC	Trilaciclib or placebo on days 1–5 of each 21-day topotecan cycle[c]

NOTE: Primary prophylaxis with G-CSF and use of ESAs were prohibited in cycle 1, although therapeutic G-CSF was allowed; after cycle 1, supportive care, including G-CSF and ESAs, was allowed as needed.
[a] Each 21-day cycle comprised IV doses of etoposide 100 mg/m^2 on days 1, 2, and 3, and carboplatin AUC 5 on day 1.
[b] Induction treatment comprised etoposide 100 mg/m^2 on days 1, 2, and 3, carboplatin AUC 5 on day 1, and atezolizumab 1200 mg on day 1 of each 21-day cycle. Maintenance treatment comprised atezolizumab 1200 mg on day 1 of each 21 day cycle; trilaciclib and placebo were not administered during the maintenance period.
[c] Topotecan 1.5 mg/m^2 was administered on days, 1, 2, 3, 4, and 5 of each 21-day cycle.
1/2/3L, first-/second-/third-line; A, atezolizumab; AUC, area under the plasma drug concentration-time curve; E/P, etoposide/carboplatin; ESA, erythropoiesis-stimulating agent; ES-SCLC, extensive-stage small cell lung cancer; G-CSF, granulocyte-colony stimulating factor.

- The treatment effect of trilaciclib was evaluated in terms of myelopreservation and antitumor efficacy
 - The primary myelopreservation endpoints were duration of severe (grade 4) neutropenia (DSN) in cycle 1, and occurrence of severe neutropenia (SN) across the treatment period
 - Secondary myelopreservation endpoints were assessed by hematopoietic lineage (neutrophils, red blood cells, and platelets)
 - Antitumor efficacy measures included objective response, progression-free survival (PFS), and overall survival (OS)
- Safety was assessed in all patients who received ≥ 1 dose of study drug (etoposide, carboplatin, atezolizumab, topotecan, or trilaciclib/placebo)

RESULTS

Patient Disposition and Baseline Characteristics

- The pooled efficacy analysis set comprised 123 and 119 patients who received trilaciclib or placebo prior to CT, respectively
- Patient demographics and baseline disease characteristics were generally comparable between treatment groups (**Table 2**)
 - Most patients were Caucasian, with an Eastern Cooperative Oncology Group performance status of 0/1, and without brain metastases

Table 2. Patient Demographics and Baseline Disease Characteristics

Parameter	Placebo prior to CT (n = 119)	Trilaciclib prior to CT (n = 123)
Median age, years (range)	64 (39–86)	64 (45–82)
Age group, n (%)		
< 65 years	61 (51.3)	66 (53.7)
≥ 65 years	58 (48.7)	57 (46.3)
Gender, n (%)		
Male	73 (61.3)	89 (72.4)
Female	46 (38.7)	34 (27.6)
Race, n (%)		
Caucasian	110 (92.4)	120 (97.6)
Other	9 (7.6)	3 (2.4)
Region, n (%)		
USA	57 (47.9)	53 (43.1)
Other	62 (52.1)	70 (56.9)
ECOG PS, n (%)		
0–1	107 (89.9)	108 (87.8)
2	12 (10.1)	15 (12.2)
Presence of brain metastases, n (%)		
Yes	28 (23.5)	27 (22.0)
No	90 (75.6)	95 (77.2)
Missing	1 (0.8)	1 (0.8)
Smoking history, n (%)		
Never smoked	8 (6.7)	7 (5.7)
Former smoker	74 (62.2)	66 (53.7)
Current smoker	36 (30.3)	49 (39.8)
Missing	1 (0.8)	1 (0.8)
Baseline LDH, n (%)		
≤ ULN	61 (51.3)	55 (44.7)
> ULN	54 (45.4)	62 (50.4)
Missing	4 (3.4)	6 (4.9)

CT, chemotherapy; ECOG PS, Eastern Cooperative Oncology Group performance status; LDH, lactate dehydrogenase; ULN, upper limit of normal.

Antitumor Efficacy of Trilaciclib Administered Prior to CT

- Tumor response rates were similar between treatment groups, with a response achieved by 56 (49.1%) and 59 (51.8%) response-evaluable patients receiving trilaciclib or placebo prior to CT, respectively (*P* = 0.7879)
- PFS and OS were comparable between treatment groups (**Figure 2**); for the trilaciclib and placebo groups:
 - Median PFS was 5.3 months (95% CI, 4.6–6.1) and 5.0 months (95% CI, 4.4–5.5), respectively (hazard ratio [HR] 0.80; 95% confidence interval [CI], 0.61–1.06; *P* = 0.1404)
 - Median OS was 10.6 months (95% CI, 9.1–11.7) and 10.6 months (95% CI, 7.9–12.8) respectively (HR 1.00; 95% CI, 0.75–1.35; *P* = 0.8136)

Figure 2. Kaplan-Meier Plot of PFS and OS in the Pooled Efficacy Analysis

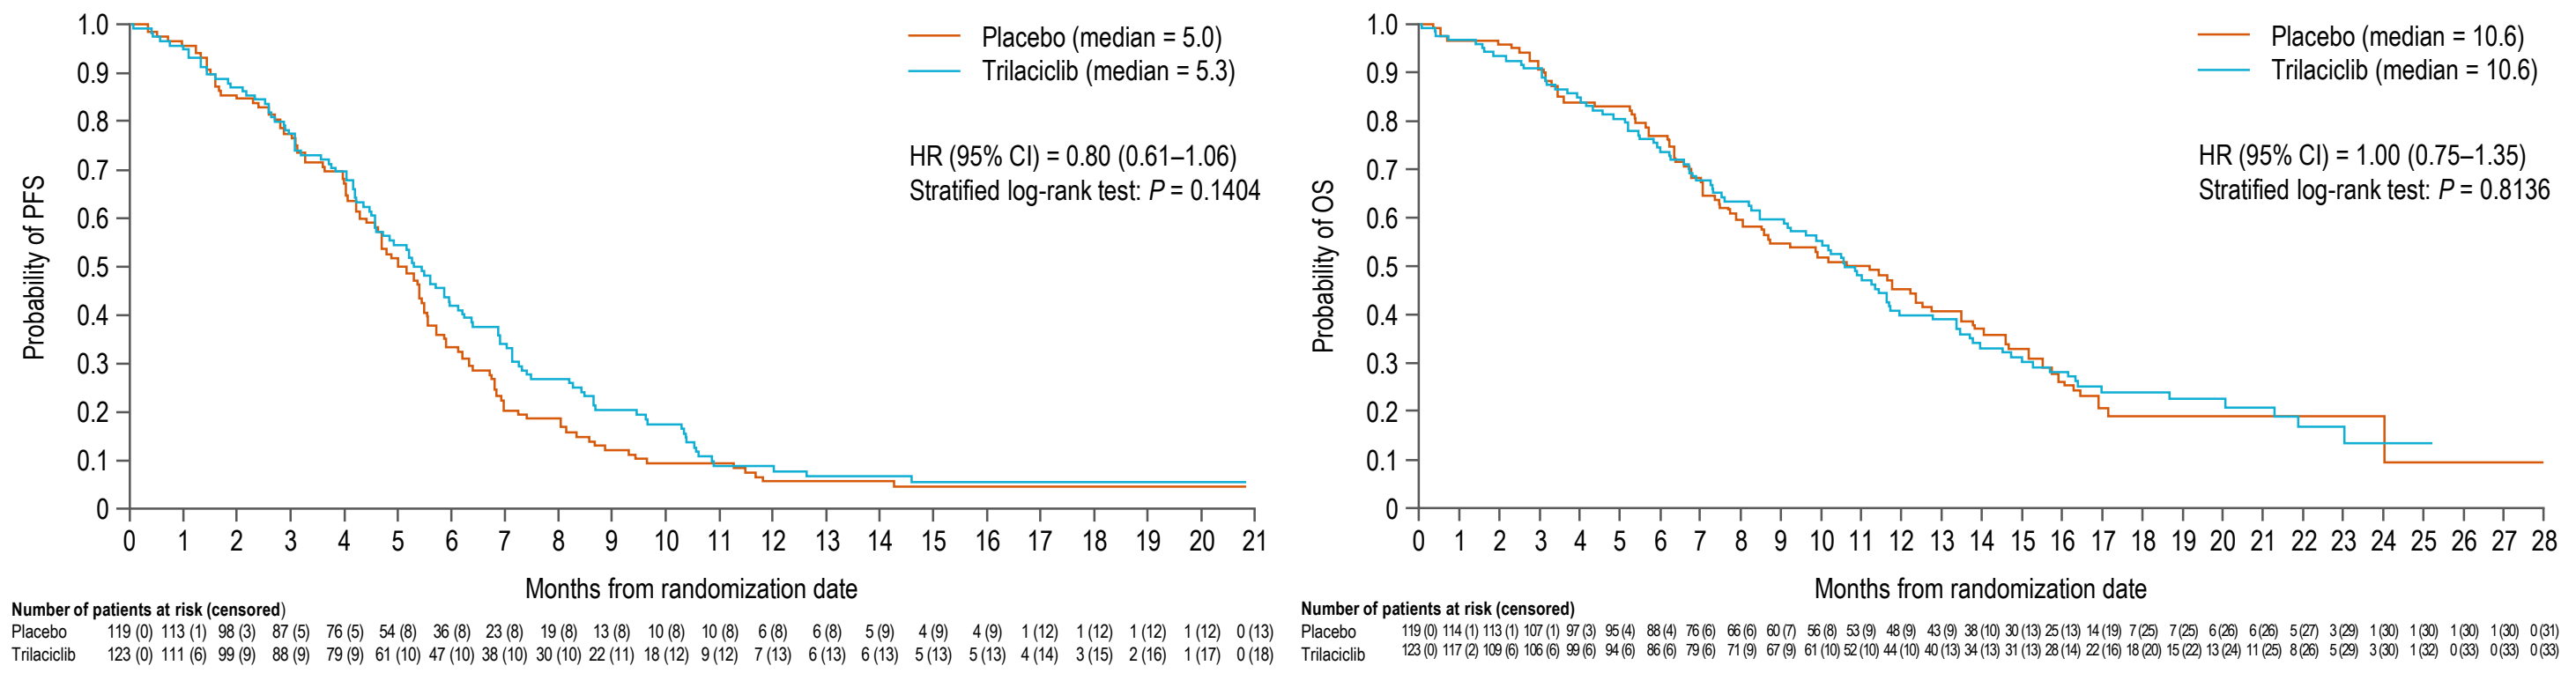

Placebo (median = 5.0)
Trilaciclib (median = 5.3)
HR (95% CI) = 0.80 (0.61–1.06)
Stratified log-rank test: P = 0.1404
Probability of PFS
Months from randomization date
Number of patients at risk (censored)
Placebo 119 (0) 113 (1) 98 (3) 87 (5) 76 (5) 54 (8) 36 (8) 23 (8) 19 (8) 13 (8) 10 (8) 10 (8) 6 (8) 6 (8) 5 (9) 4 (9) 4 (9) 1 (12) 1 (12) 1 (12) 0 (13)
Trilaciclib 123 (0) 111 (6) 99 (9) 88 (9) 79 (9) 61 (10) 47 (10) 38 (10) 30 (10) 22 (11) 18 (12) 9 (12) 7 (13) 6 (13) 6 (13) 5 (13) 5 (13) 4 (14) 3 (15) 2 (16) 1 (17) 0 (18)
Placebo (median = 10.6)
Trilaciclib (median = 10.6)
HR (95% CI) = 1.00 (0.75–1.35)
Stratified log-rank test: P = 0.8136
Probability of OS
Months from randomization date
Number of patients at risk (censored)

CI, confidence interval; HR, hazard ratio; OS, overall survival; PFS, progression-free survival.

Myelopreservation Efficacy of Trilaciclib Administered Prior to CT

- Addition of trilaciclib prior to CT significantly decreased most measures of multilineage CIM and the need for supportive care interventions (**Figure 1**)
 - The primary endpoints of DSN in cycle 1 (a surrogate for febrile neutropenia and infections), and occurrence of SN were both significantly reduced with trilaciclib versus placebo
 - Mean (standard deviation) DSN was 0 (1.8) days with trilaciclib versus 4 (5.1) days with placebo (*P* < 0.0001)

Figure 1. Myelopreservation Endpoints in the Pooled Efficacy Analysis

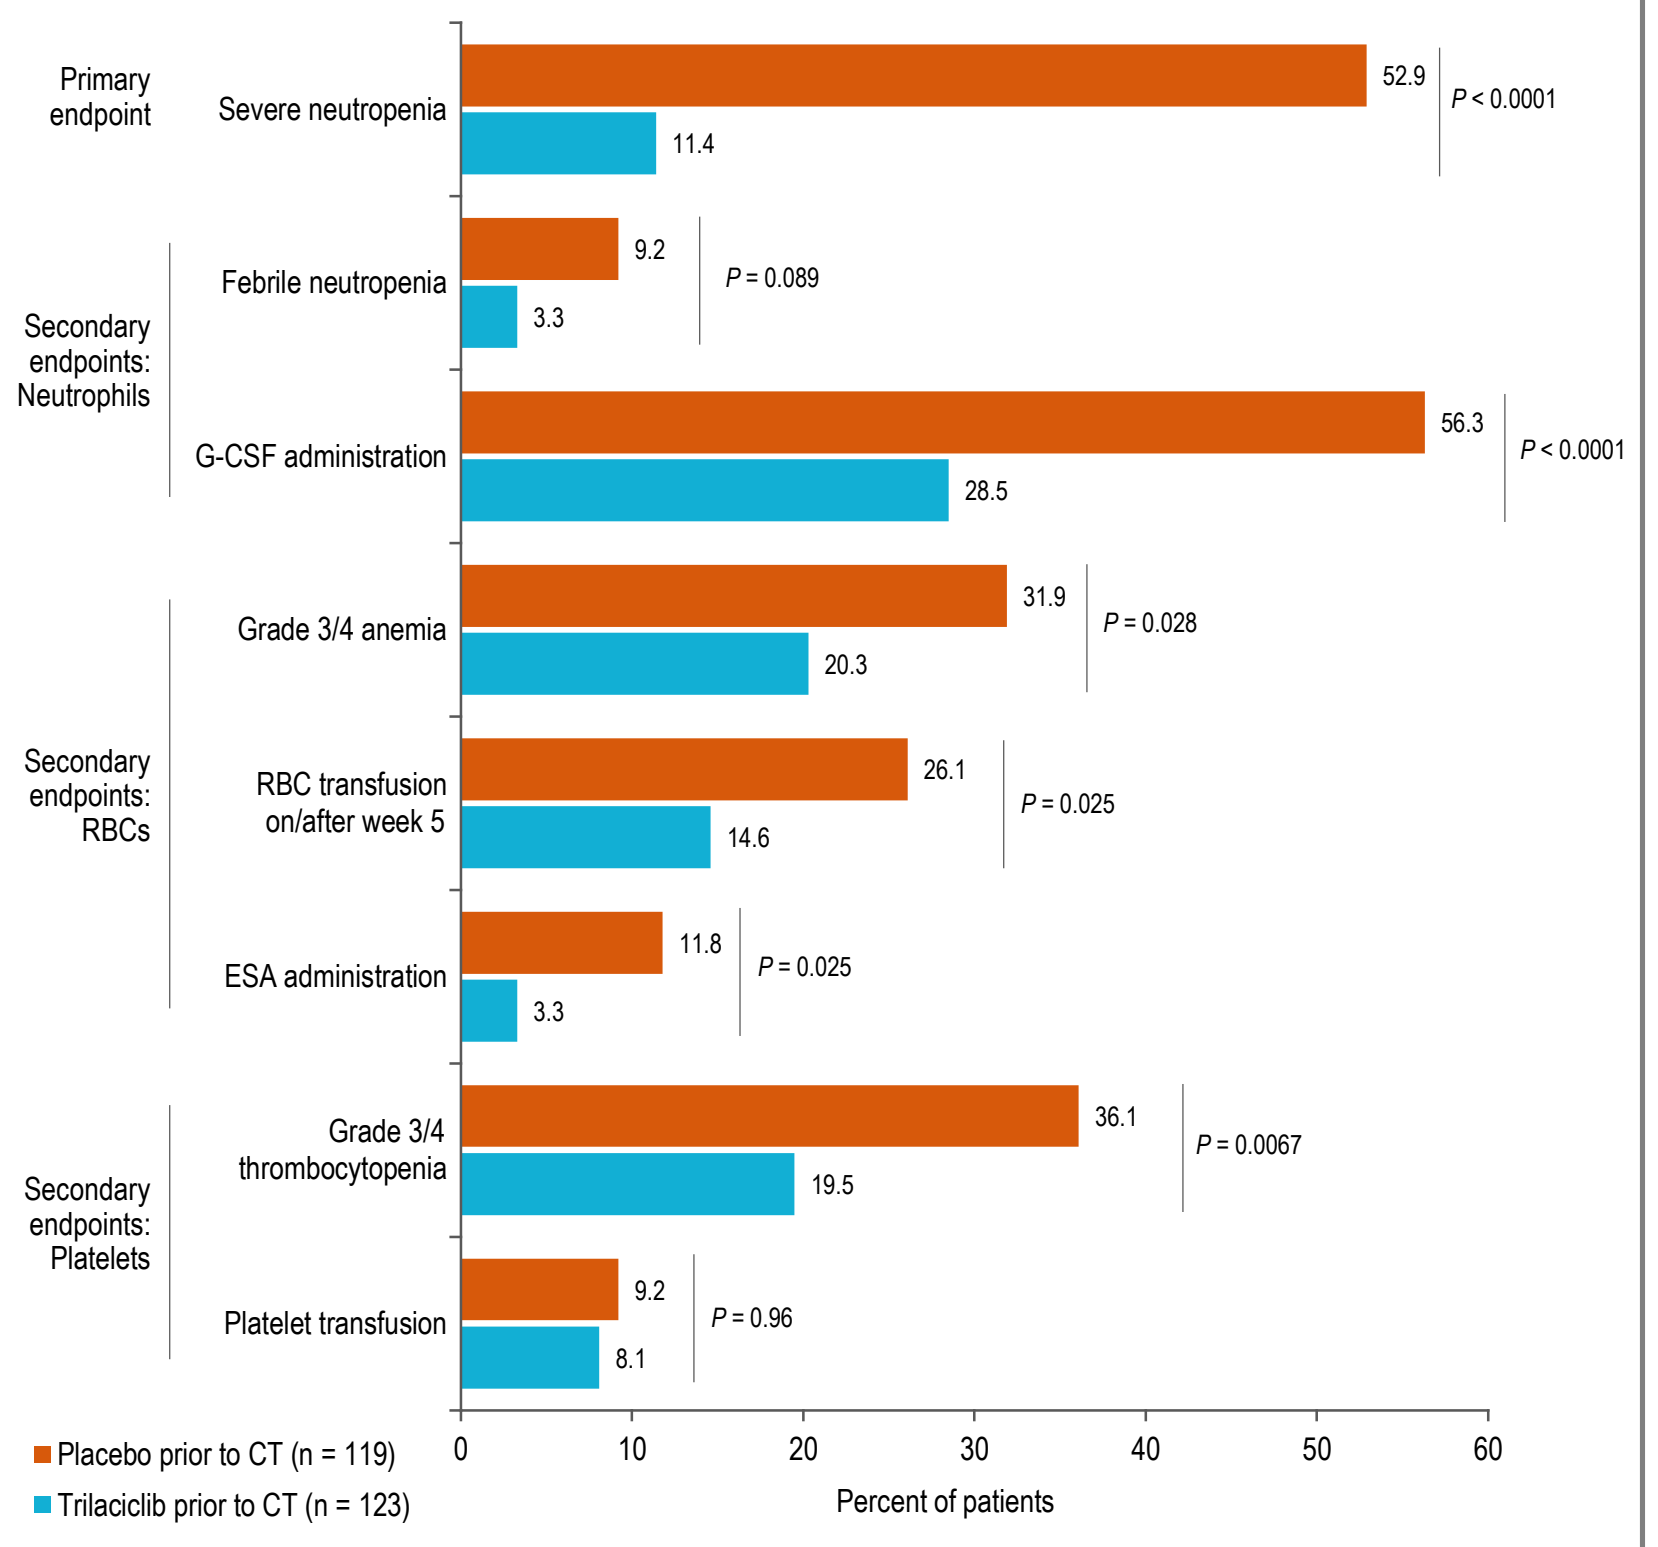

Primary endpoint
Severe neutropenia 52.9 / 11.4 P < 0.0001
Secondary endpoints: Neutrophils
Febrile neutropenia 9.2 / 3.3 P = 0.089
G-CSF administration 56.3 / 28.5 P < 0.0001
Secondary endpoints: RBCs
Grade 3/4 anemia 31.9 / 20.3 P = 0.028
RBC transfusion on/after week 5 26.1 / 14.6 P = 0.025
ESA administration 11.8 / 3.3 P = 0.025
Secondary endpoints: Platelets
Grade 3/4 thrombocytopenia 36.1 / 19.5 P = 0.0067
Platelet transfusion 9.2 / 8.1 P = 0.96
Placebo prior to CT (n = 119)
Trilaciclib prior to CT (n = 123)
Percent of patients

CT, chemotherapy; ESA, erythropoiesis-stimulating agent; G-CSF, granulocyte-colony stimulating factor; RBC, red blood cell.

Additional Clinical Benefits of Trilaciclib Administered Prior to CT

- Among patients who continued after cycle 1, 11 patients (8.9%) receiving trilaciclib prior to CT required ≥ 1 CT dose reduction versus 36 patients (30.3%) receiving placebo (event rate per 100 cycles: 2.8 vs 9.3, respectively)
- Fewer patients receiving trilaciclib had infection serious adverse events, or received IV antibiotics, compared with those receiving placebo (6.5% vs 10.1%, and 19.5% vs 23.5%, respectively)

Safety and Tolerability

- A summary of treatment-emergent adverse events (TEAEs) among patients receiving trilaciclib or placebo prior to CT is provided in **Table 3**
- The most frequently reported TEAEs (in ≥ 20% of patients in both treatment groups) were neutropenia, anemia, thrombocytopenia, nausea, and fatigue
- Significantly fewer patients receiving trilaciclib prior to CT had high-grade hematologic toxicities compared with patients receiving placebo

Table 3. Incidence of TEAEs (Occurring in ≥ 20% of Patients) and Summary of High-Grade Hematologic Toxicities in the Pooled Safety Analysis

Event, n (%)	Placebo prior to CT (n = 118)		Trilaciclib prior to CT (n = 122)	
Any TEAE	114 (96.6)		115 (94.3)	
Any placebo-/trilaciclib-related TEAE	49 (41.5)		45 (36.9)	
Any serious TEAE	30 (25.4)		36 (29.5)	
Any placebo-/trilaciclib-related serious TEAE	1 (0.8)		2 (1.6)	
Any TEAE leading to study drug discontinuation	13 (11.0)		11 (9.0)	
Any TEAE leading to death	3 (2.5)		6 (4.9)	
Adverse event of special interest[a]	10 (8.5)		23 (18.9)	
Most-common TEAEs (occurring in ≥ 20% of patients)[b]				
Neutropenia	78 (66.1)		51 (41.8)	
Anemia	71 (60.2)		46 (37.7)	
Thrombocytopenia	50 (42.4)		37 (30.3)	
Nausea	39 (33.1)		41 (33.6)	
Fatigue	32 (27.1)		41 (33.6)	
Alopecia	30 (25.4)		16 (13.1)	
Leukopenia	28 (23.7)		10 (8.2)	
High-grade hematologic toxicities, n (%)	**Grade 3 or 4**	**Grade 4**	**Grade 3 or 4**	**Grade 4**
Any hematologic TEAE	91 (77.1)	62 (52.5)	54 (44.3)	19 (15.6)
Neutropenia	81 (68.6)	58 (49.2)	39 (32.0)	10 (8.2)
Thrombocytopenia	39 (33.1)	21 (17.8)	22 (18.0)	10 (8.2)
Anemia	40 (33.9)	1 (0.8)	20 (16.4)	0 (0)
Leukopenia	20 (16.9)	5 (4.2)	5 (4.1)	1 (0.8)
Febrile neutropenia	11 (9.3)	5 (4.2)	4 (3.3)	3 (2.5)
Lymphopenia	1 (0.8)	0 (0)	1 (0.8)	0 (0)

[a] Most commonly grade 1 or 2 injection site reactions and phlebitis/thrombophlebitis.
[b] Occurring in ≥ 20% of patients in either treatment group, ordered from highest to lowest frequency in the placebo group.
CT, chemotherapy; TEAE, treatment-emergent adverse event.

CONCLUSIONS

- Addition of trilaciclib prior to CT significantly and meaningfully reduced both CIM and its consequences, was associated with a substantial reduction in high-grade hematologic TEAEs, and had no detrimental effects on PFS or OS
- Trilaciclib reduces the toxicity of CT in ES-SCLC, and has the potential to become a new standard of care for preventing CIM in this patient population

REFERENCES

1. Weiss JM, et al. *Ann Oncol.* 2019;30:1613–21.
2. Daniel D, et al. *Ann Oncol.* 2019;30(5 suppl):v713.
3. Hart LL, et al. *J Clin Oncol.* 2019;37(15 suppl):8505.

ACKNOWLEDGMENTS

- We thank all of the investigators and site staff, with special thanks to the patients and their families, for their participation in the studies
- Medical writing assistance was provided by Alligent Europe (Envision Pharma Group), funded by G1 Therapeutics, Inc.

Corresponding author: Jared Weiss (jared_weiss@med.unc.edu)

Presented at the American Society of Clinical Oncology (ASCO) Virtual Meeting 2020, May 29–31, 2020

Disclaimer: Copies of this poster obtained through QR (Quick Response) and/or text key codes are for personal use only and may not be reproduced without written permission of the authors